CIDP Disease Burden — Results of a US Nationwide Patient Survey

Jeffrey A. Allen,[1] Lisa Butler,[2] Todd D. Levine,[3] Ann L. Bullinger[4] and Carol L. Koski[2]

[1]Department of Neurology, University of Minnesota, Minneapolis, MN, USA.
[2]GBS|CIDP Foundation International, Conshohocken, PA, USA.
[3]Phoenix Neurological Associates, Phoenix, AZ, USA.
[4]CSL Behring, King of Prussia, PA, USA.

Introduction

- Chronic inflammatory demyelinating polyneuropathy (CIDP) is a rare peripheral neuropathy characterized by proximal upper and lower limb weakness, and sensory and reflex loss[1,2]

CIDP DISEASE BURDEN

- CIDP is most frequently treated with intravenous immunoglobulin (IVIg) or corticosteroids; but patients may still report symptoms such as pain, fatigue, and a multitude of personal and professional limitations[3–5]
- Treatment with IVIg commonly requires lengthy infusions which can result in missing time from work/school

US PATIENT SURVEY

- A nationwide survey of US CIDP patients was conducted to assess the impact of disease-related disability and treatment on domestic arrangements, and work activities

Aim

- To assess the impact of CIDP disease-related disability and treatment on domestic arrangements and work activities through a nationwide survey of US CIDP patients

Methods

- Approximately 3250 individuals aged ≥18 years, recruited by the GBS|CIDP Foundation and self-reported to have CIDP, were invited to complete an online survey; of these, 475 completed the survey and their responses were used to assess disease and treatment burden
- Data were analyzed overall and by stratification of patients based on the likelihood of an accurate CIDP diagnosis, defined as shown in **Table 1**

Table 1: Stratification of patients based on likelihood of accurate CIDP diagnosis

Unlikely CIDP patient	• Reported no muscle weakness as symptom of CIDP • Did not report having neurophysiologic tests performed when diagnosed
Somewhat likely CIDP patient	• Reported weakness, but not consistently • Reported symptoms were at their worst in less than 2 months (without prior diagnosis of Guillain-Barré syndrome [GBS]) • Reported symptoms were not symmetric
Likely CIDP patient	• Absence of the above-listed factors • Includes patients whose symptoms reached their worst in less than 2 months with a previous diagnosis of GBS

- Overall results as well as any deviation from overall results by stratification are presented
- There were 187, 170 and 118 patients with likely, somewhat likely and unlikely CIDP, respectively (**Figure 1**)

Figure 1: Percentage of respondents classified as either likely, somewhat likely or unlikely CIDP patients

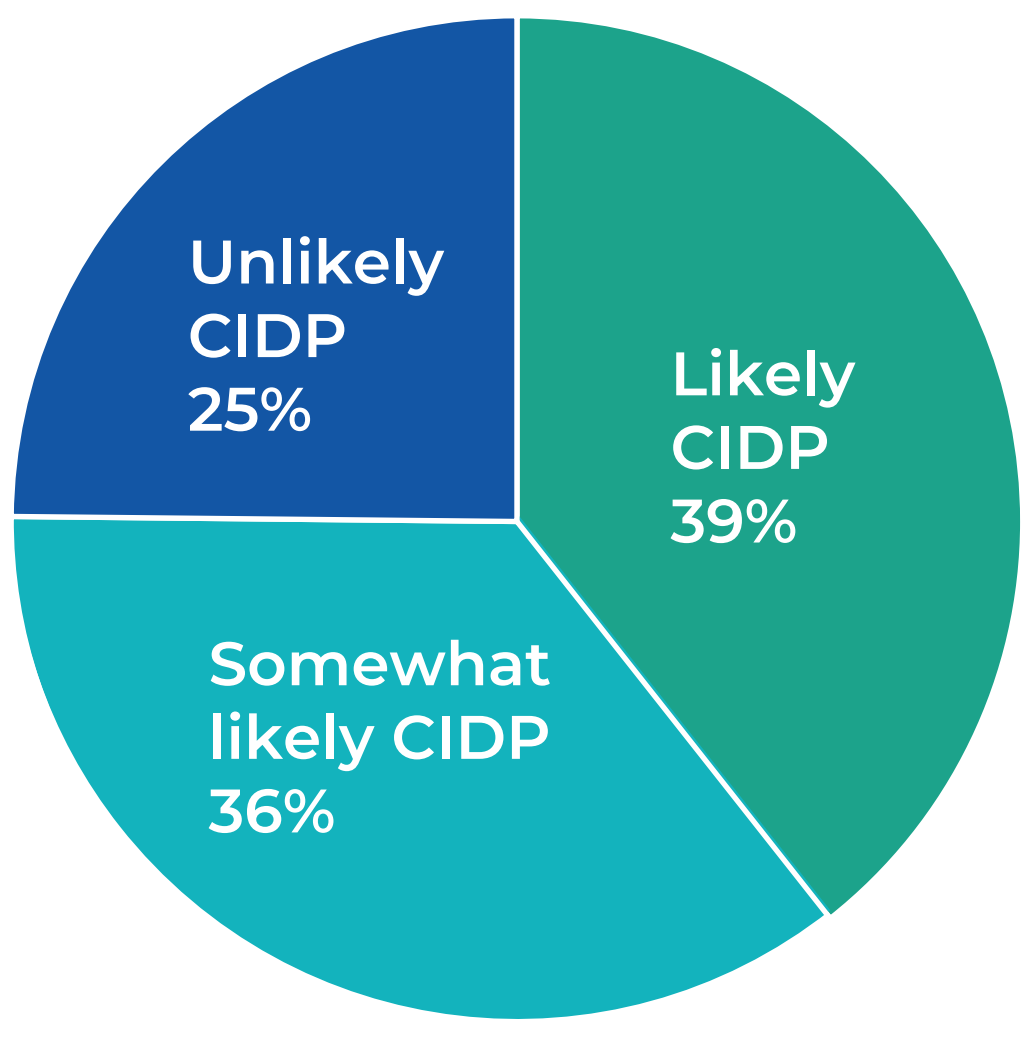
Unlikely CIDP 25%
Likely CIDP 39%
Somewhat likely CIDP 36%

Patients were classified according to the criteria listed in Table 1.

Results

SYMPTOMS BY LIKELIHOOD OF CIDP DIAGNOSIS

- The number of CIDP symptoms ever experienced stratified by likelihood of CIDP diagnosis is shown in **Figure 2**
 - Overall, patients experienced around 9 symptoms with almost half having experienced all 10 symptoms queried
 - Those who were unlikely to have received an accurate CIDP diagnosis typically had fewer symptoms versus patients with likely CIDP

Figure 2: Symptoms by likelihood of CIDP diagnosis

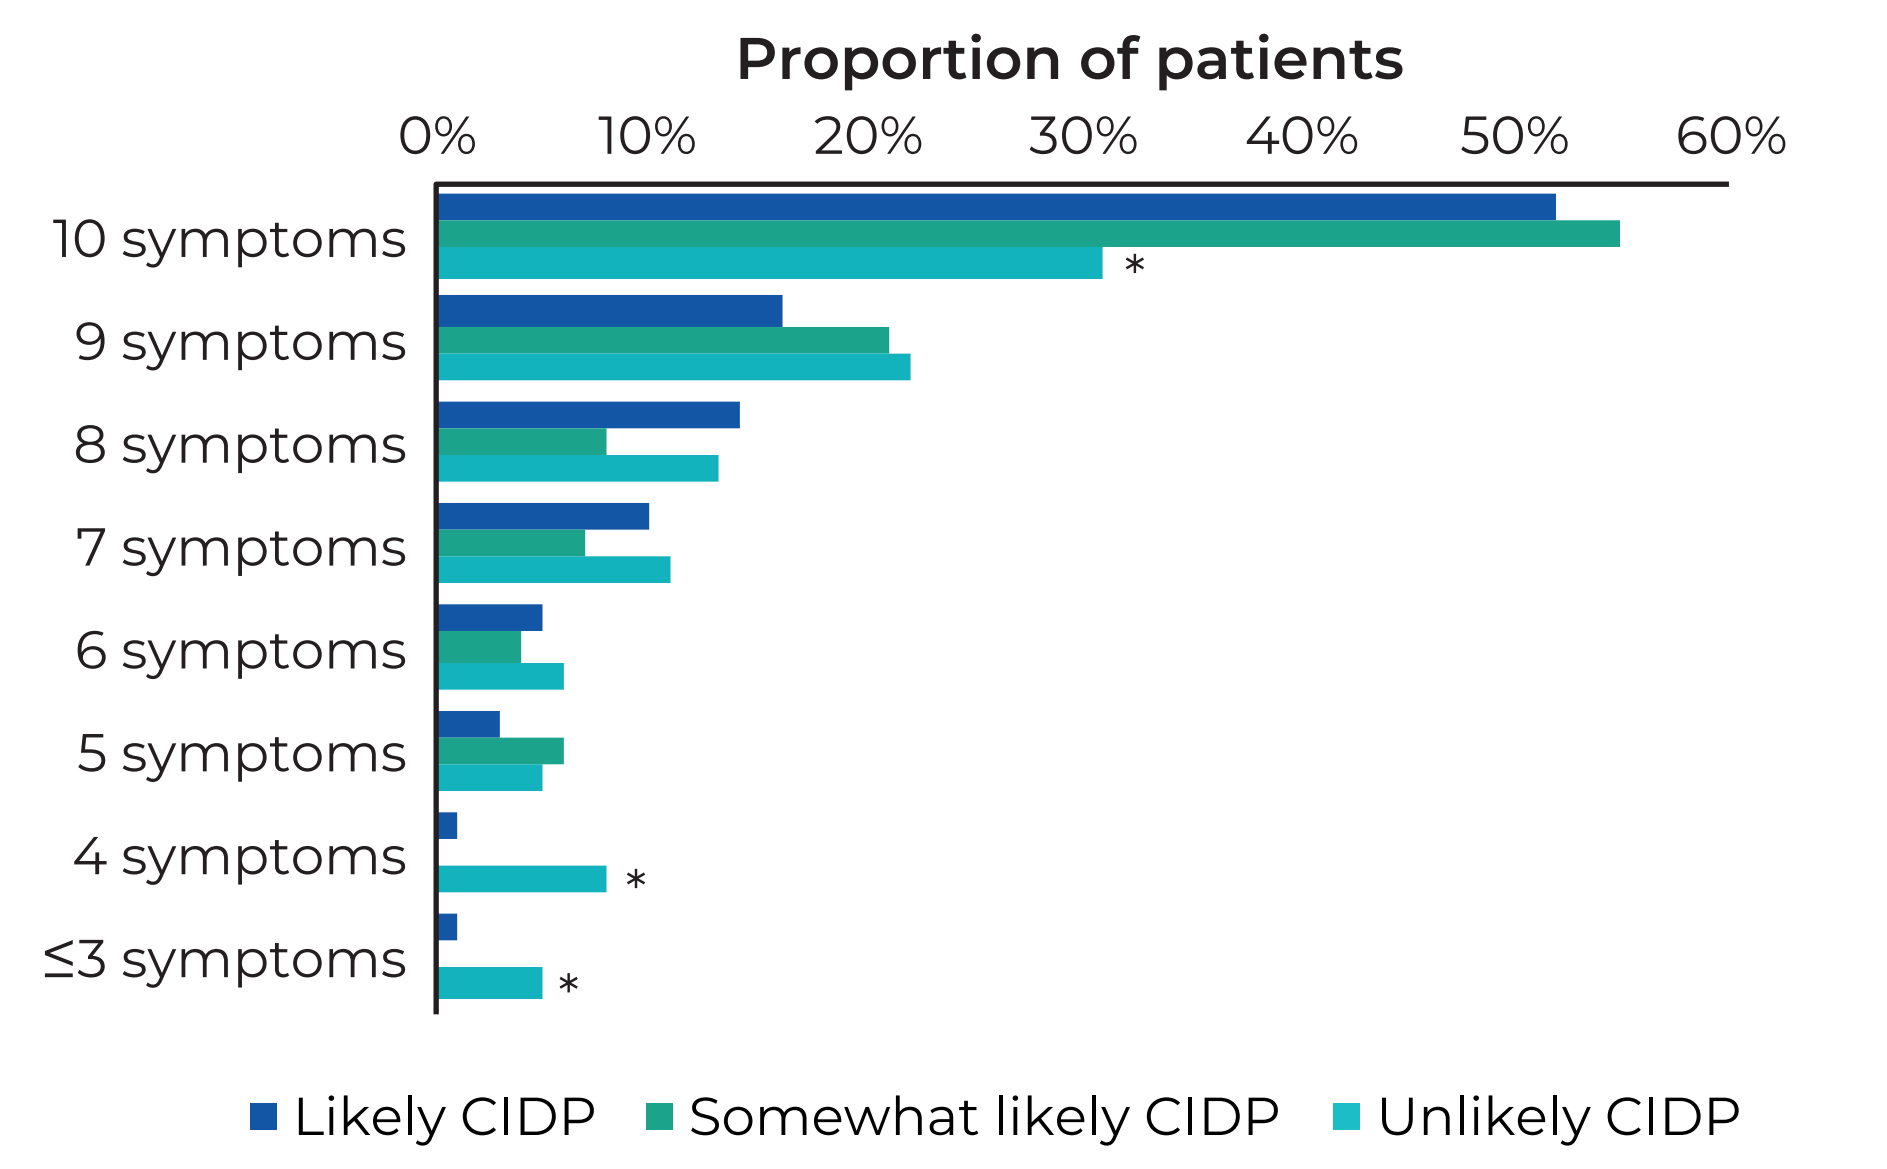
Proportion of patients
0%
10%
20%
30%
40%
50%
60%
10 symptoms
9 symptoms
8 symptoms
7 symptoms
6 symptoms
5 symptoms
4 symptoms
≤3 symptoms
*
Likely CIDP
Somewhat likely CIDP
Unlikely CIDP

Shows the amount of symptoms experienced by patients stratified by likelihood of accurate diagnosis. Patients were asked to select which of 10 CIDP symptoms applied to them (**1.** Weakness in shoulders/arms; **2.** Weakness in hips/legs; **3.** Heaviness in legs; **4.** Weakness in hands/fingers; **5.** Weakness in feet/toes; **6.** Numbness in hands or feet; **7.** Tingling; **8.** Pain [for example feeling of burning, or electrical sensation]; **9.** Loss of balance/coordination; **10.** Fatigue). *p<0.05 vs. other groups.

MOST BOTHERSOME SYMPTOMS

- The most bothersome symptoms at the time of starting treatment stratified by likelihood of CIDP diagnosis are shown in **Figure 3**

Figure 3: Most bothersome symptoms at the time of starting medication

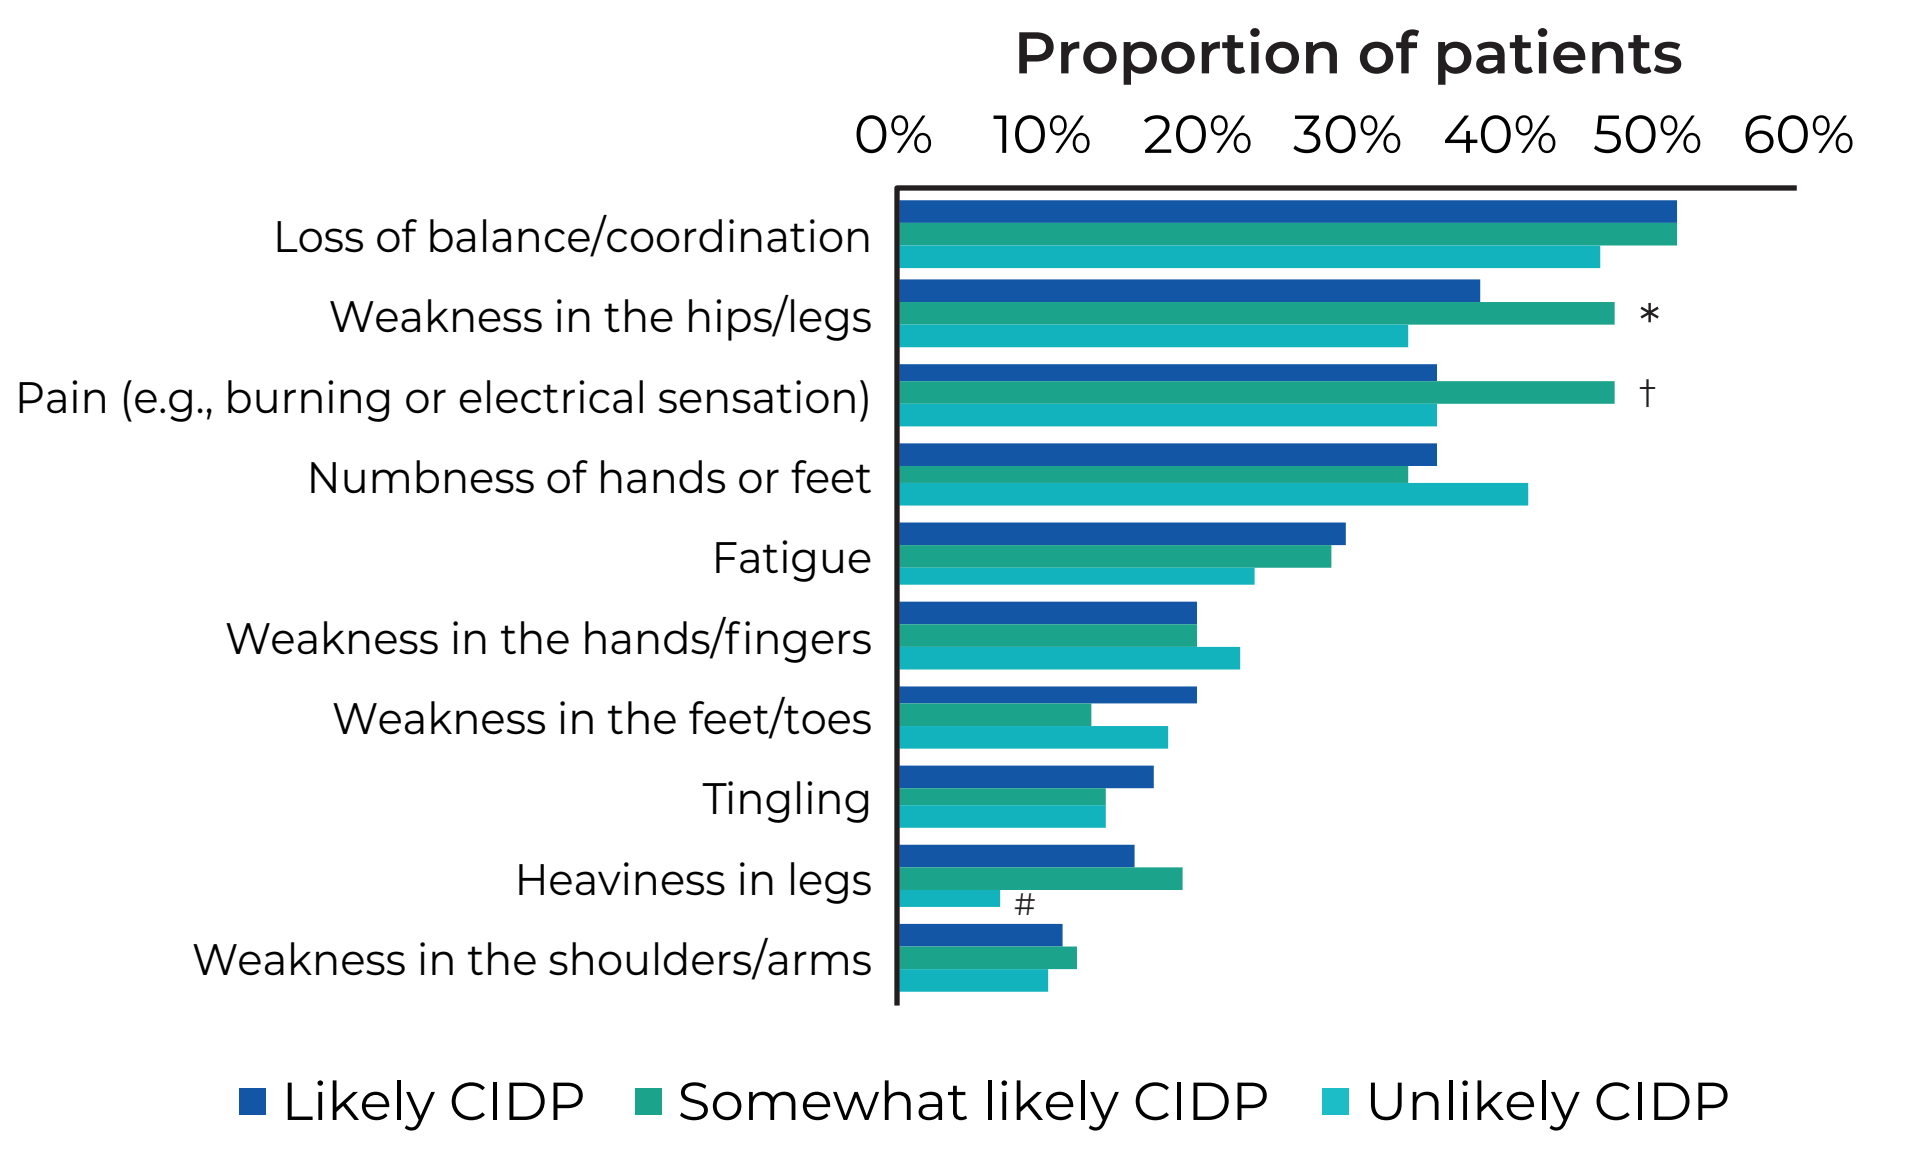
Proportion of patients
0%
10%
20%
30%
40%
50%
60%
Loss of balance/coordination
Weakness in the hips/legs
Pain (e.g., burning or electrical sensation)
Numbness of hands or feet
Fatigue
Weakness in the hands/fingers
Weakness in the feet/toes
Tingling
Heaviness in legs
Weakness in the shoulders/arms
*
†
#
Likely CIDP
Somewhat likely CIDP
Unlikely CIDP

Patients could select up to 3 symptoms as most bothersome. *p<0.05 vs. "unlikely CIDP"; †p<0.05 vs. "likely CIDP"; #p<0.05 vs. "somewhat likely" and "likely CIDP".

GENERAL DISEASE BURDEN

- A summary of the impact of CIDP on professional life, living circumstances, mobility and quality of life is shown in **Figure 4**

Figure 4: Summary of general CIDP disease burden

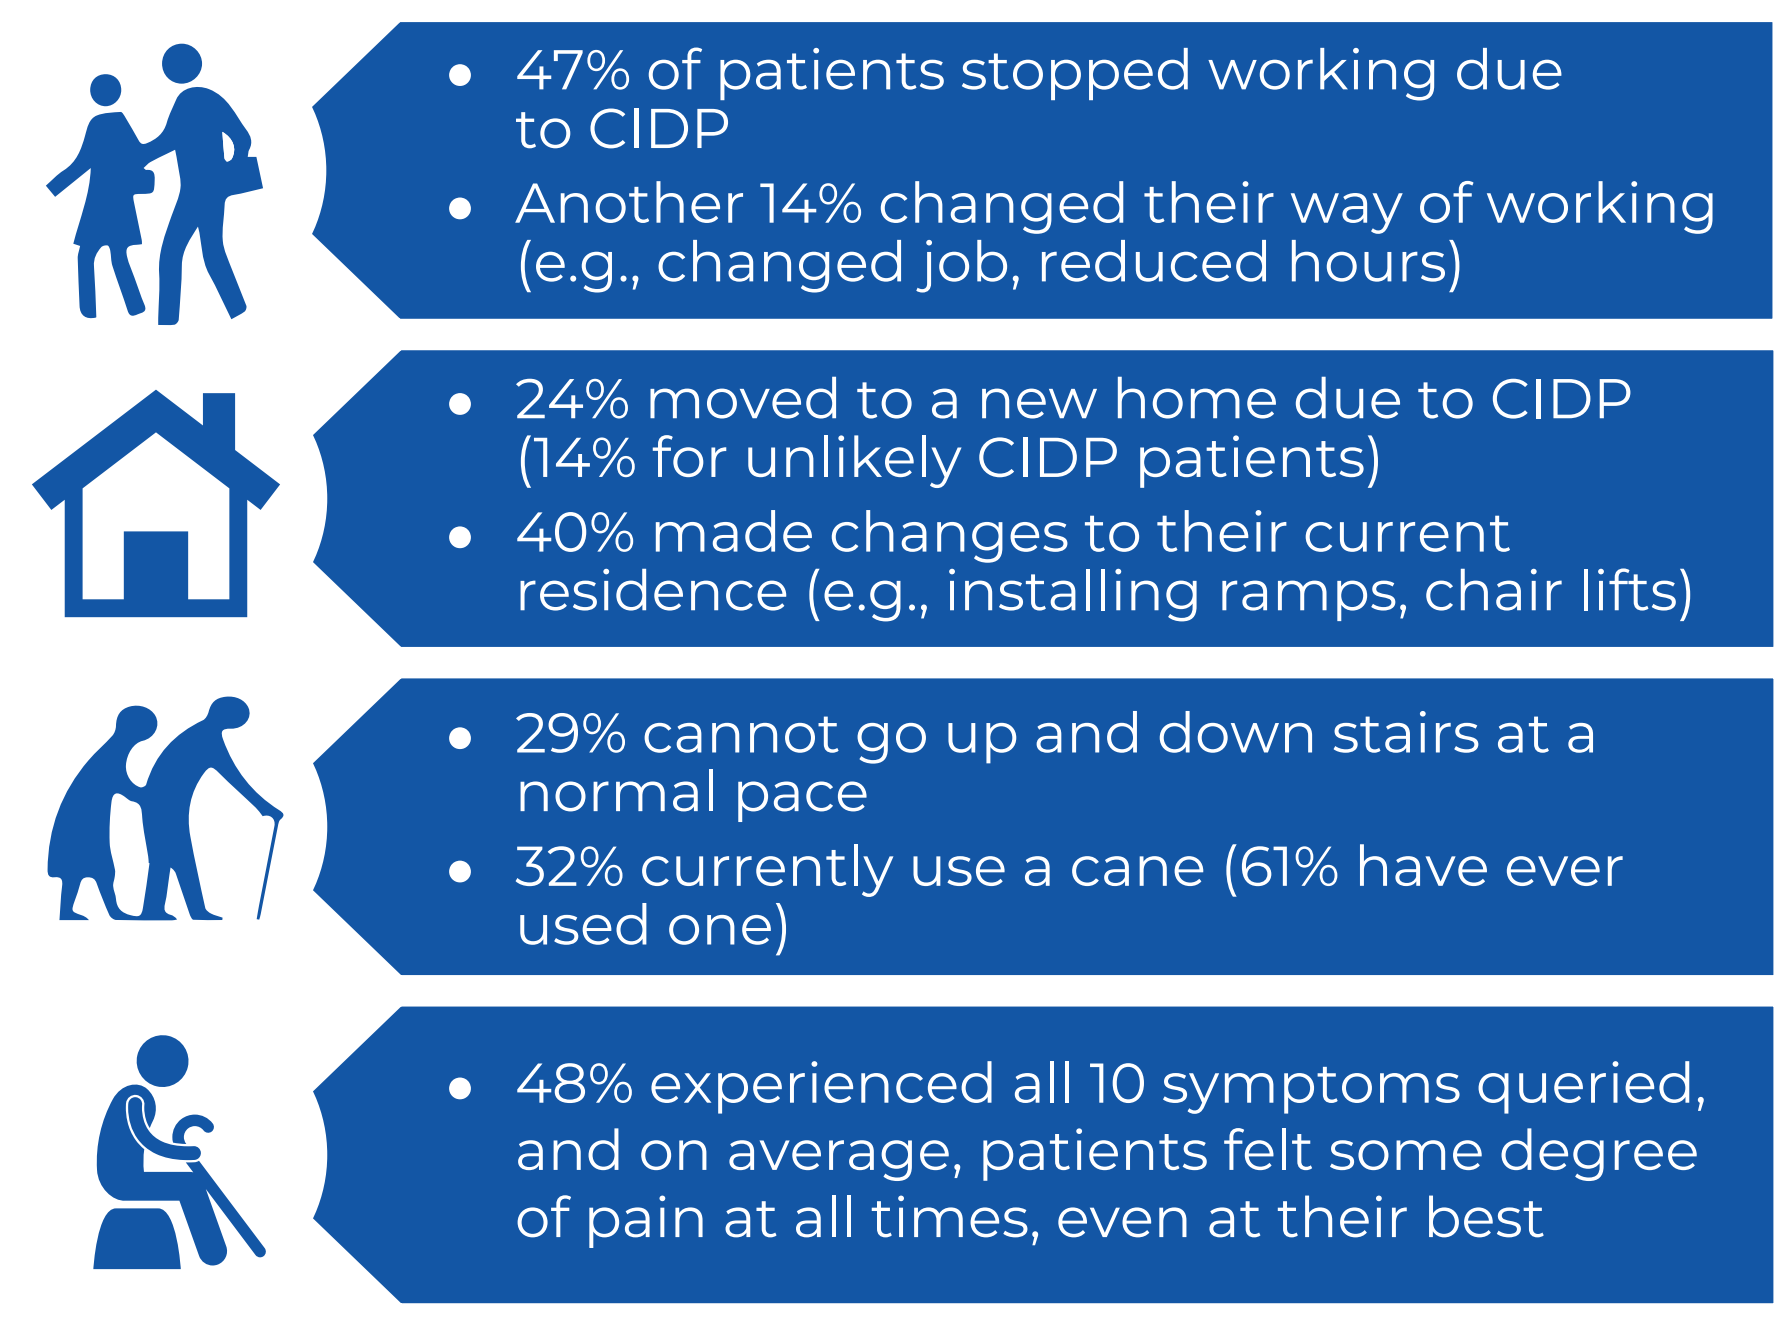
• 47% of patients stopped working due to CIDP
• Another 14% changed their way of working (e.g., changed job, reduced hours)
• 24% moved to a new home due to CIDP (14% for unlikely CIDP patients)
• 40% made changes to their current residence (e.g., installing ramps, chair lifts)
• 29% cannot go up and down stairs at a normal pace
• 32% currently use a cane (61% have ever used one)
• 48% experienced all 10 symptoms queried, and on average, patients felt some degree of pain at all times, even at their best

IMPACT ON WORK/SCHOOL

- Overall, 20% of respondents reported missing time from work/school due to CIDP symptoms; **Figure 5A** shows these 20% stratified by number of days missed
- Of respondents currently on IVIg therapy, 28% reported missing time from work/school due to infusions; **Figure 5B** shows these 28% stratified by number of days missed

Figure 5: Time missed from work/school in the past month

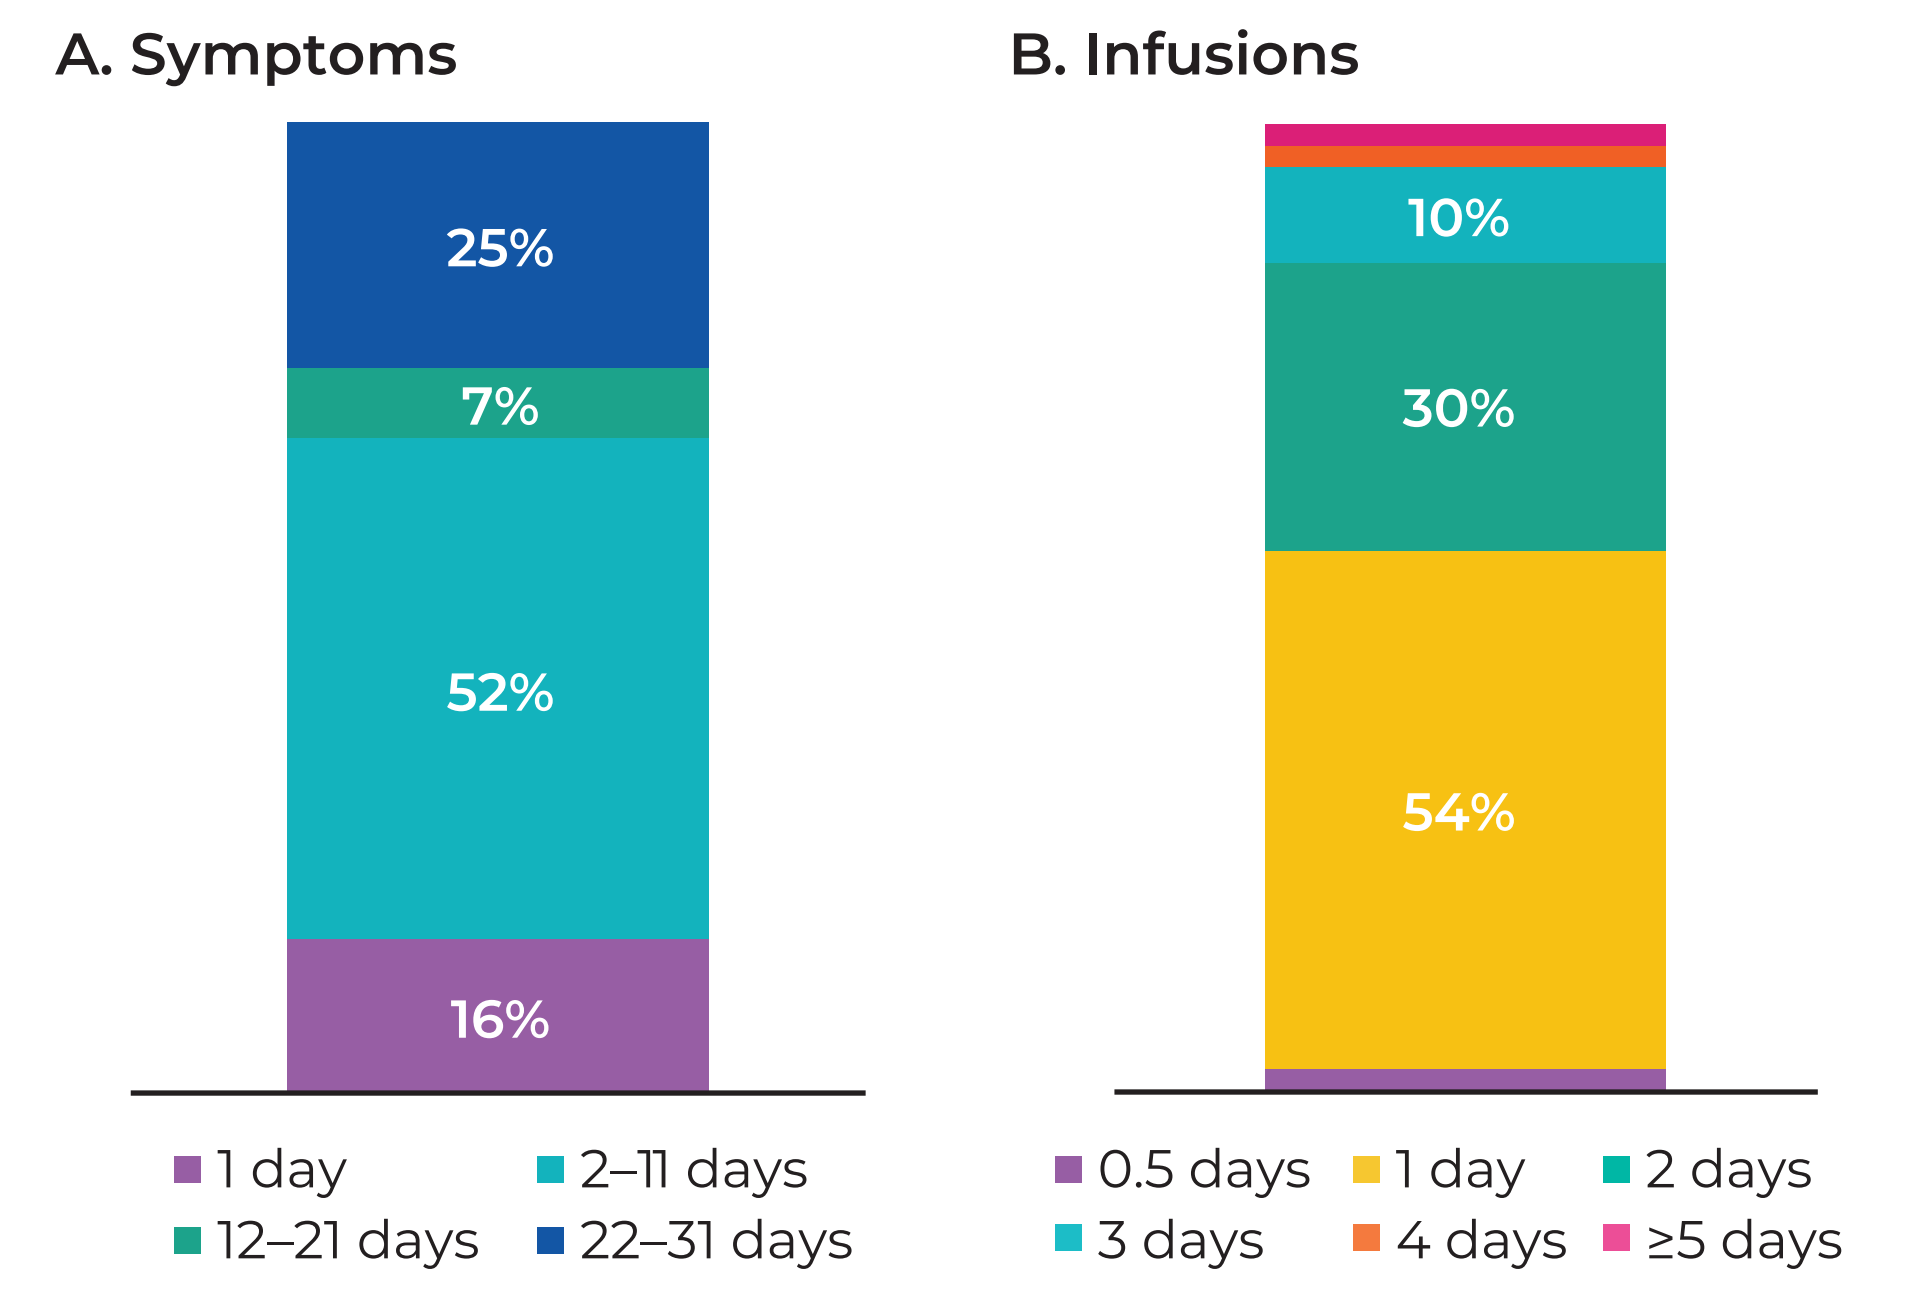
A. Symptoms
25%
7%
52%
16%
1 day
2–11 days
12–21 days
22–31 days
B. Infusions
10%
30%
54%
0.5 days
1 day
2 days
3 days
4 days
≥5 days

The amount of time missed from work/school in the past month due to **A.** symptoms (among those reporting missing any time) and **B.** IVIg infusions (among patients on IVIg who reported missing any time).

TREATMENT BURDEN

- The two most common treatments were IVIg (63% currently using; 93% have ever used) and corticosteroids (19% currently using; 59% have ever used)
- Tolerability: A higher proportion of patients discontinued corticosteroids for poor tolerability compared to IVIg (**Figure 6**)
- Among patients discontinuing treatment, a higher proportion discontinued IVIg due to difficulty with administering the treatment vs with corticosteroids (**Figure 6**)
 - A subgroup analysis (n=32) showed that benefits of subcutaneous immunoglobulin (SCIg) administration and avoiding difficulties with IVIg infusion were the most common reasons for SCIg preference (81% and 38% respectively)

Figure 6: Primary reason for treatment discontinuation

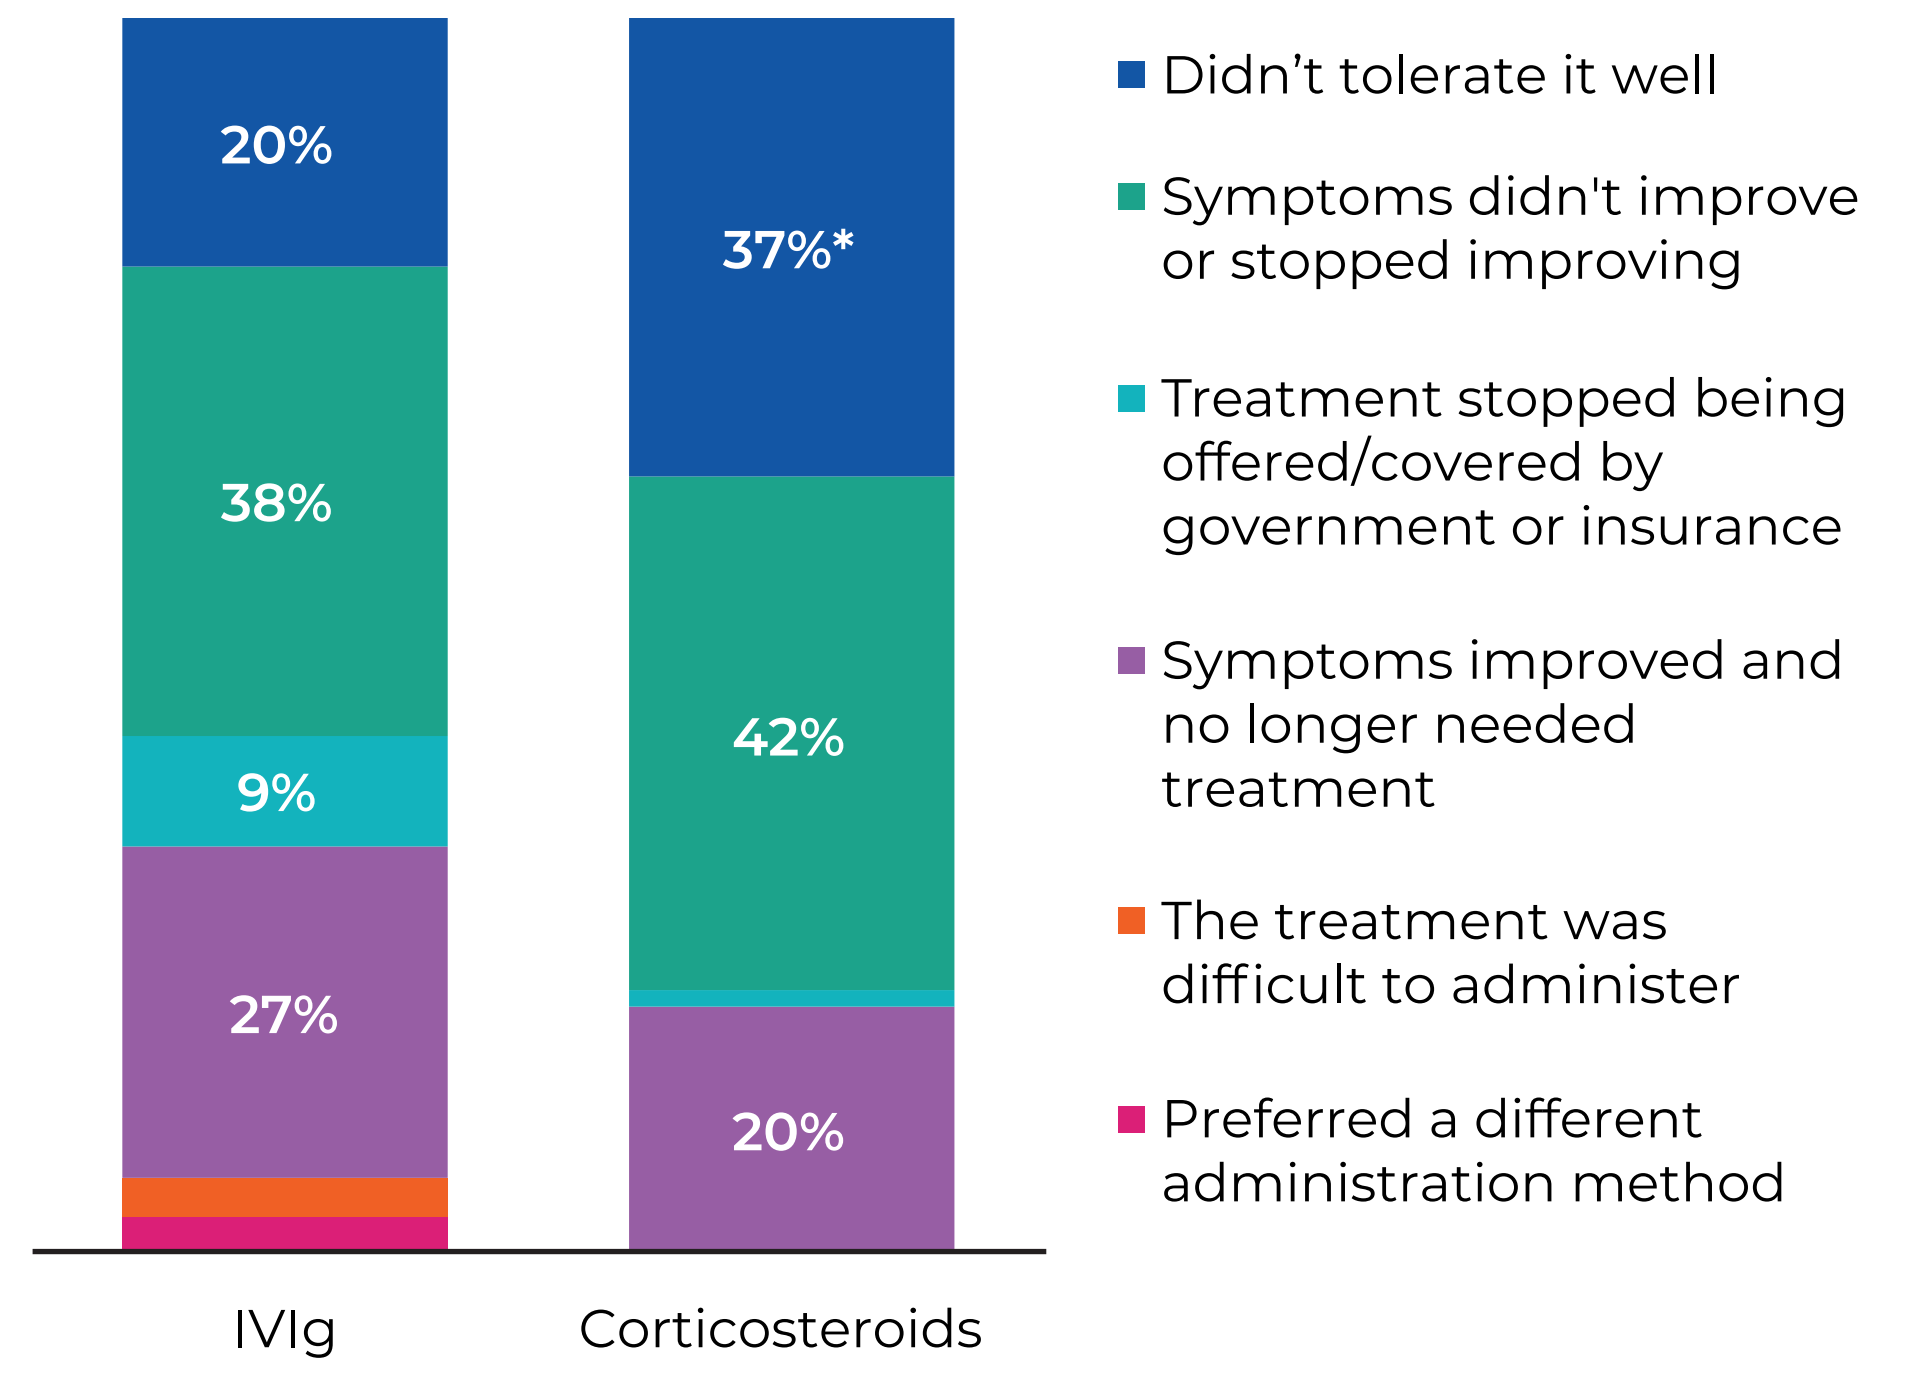
20%
38%
9%
27%
37%*
42%
20%
IVIg
Corticosteroids
Didn't tolerate it well
Symptoms didn't improve or stopped improving
Treatment stopped being offered/covered by government or insurance
Symptoms improved and no longer needed treatment
The treatment was difficult to administer
Preferred a different administration method

Primary reason for treatment discontinuation among users of IVIg and corticosteroids who have discontinued either treatment. *p<0.05 vs. other treatment.

Conclusion

- The results from this US nationwide survey demonstrate that both CIDP and commonly prescribed CIDP treatments are associated with disease and treatment burdens that impact school, work and home activities

References

1. Mathey EK, et al. *J Neurol Neurosurg Psychiatry*. 2015; **86**:973–985.
2. Dalakas MC. *Nat Rev Neurol*. 2011; **7**:507–517.
3. Santos PL, et al. *Arq Neuropsiquiatr*. 2014; **72**:179–183.
4. Merkies IS, et al. *Eur Neurol*. 2016; **75**:199–206.
5. Merkies IS, et al. *Neurology*. 1999; **53**:1648–1654.

Editorial assistance was provided by Meridian HealthComms.

Presented at the 2018 Peripheral Nerve Society (PNS) Annual Meeting, Baltimore, MD, USA; July 22–25, 2018

CSL Behring