Exploring Patient Treatment Experience with Subcutaneous C1INH Prophylaxis for Hereditary Angioedema: Impact on Breakthrough Attacks, Quality of Life, and Rescue Medication Use

Licensed by COPYRIGHTAGENCY
You must not copy this work without permission
+612 9394 7600
copyright.com.au

Lumry W[1], Craig T[2], Anderson J[3], Riedl M[4], Li H[5], Tachdjian R[6], Manning M[7], Bajcic P[8], Wang S[9], Bellenger MA[9], Koochaki P[9], Rodino F[10], and Bernstein JA[11, 12]

[1]Allergy & Asthma Specialists of Dallas, Dallas, TX [2]Penn State University, Allergy Asthma and Immunology, Departments of Medicine, Pediatrics, Biomedical Sciences, Hershey, PA [3]AllerVie Health, Birmingham, AL [4]Division of Rheumatology, Allergy & Immunology, University of California – San Diego, La Jolla, CA [5]Institute for Asthma & Allergy, Chevy Chase, MD [6]Pediatric Allergy and Immunology, UCLA School of Medicine, Los Angeles, CA [7]Allergy, Asthma & Immunology Associates, Ltd, Scottsdale, AZ [8]CSL Behring, King of Prussia, PA [9]ICON plc, Blue Bell, PA [10]Churchill Outcomes Research, LLC, Red Bank, NJ [11]Division of Rheumatology, Allergy and Immunology University of Cincinnati, Cincinnati, OH [12]Bernstein Allergy Group, Inc., Cincinnati, OH

CONCLUSIONS

- This research study utilized real world evidence via qualitative patient interviews to assess the disease experiences, treatment patterns, and impacts on people living with HAE.
- Study findings support that LTP with C1INH(SC) is an effective and well-tolerated option for HAE patients, leading to improved disease control and enhanced QOL, and a reduction in on-demand medication usage.
- Limitations of the study include a relatively small cohort sample size, possible selection bias, and a potential recall bias from retrospective interviews.

BACKGROUND

- The symptomatic and disease burden of hereditary angioedema (HAE) impacts the gamut of health-related quality of life (QOL) for patients and their families, including physical, emotional, and social functioning, education and work productivity, and social interactions.[1-4]
- Given that treatment goals for HAE include achieving total control of the disease and normalizing patients' QOL, long-term prophylaxis (LTP) with medication is currently the primary way to prevent attacks and reduce the disease burden.[5,6]
- Subcutaneous C1INH replacement therapy (C1INH[SC]; HAEGARDA®) is a first-line option for LTP, self-administered by patients/caregivers, with on-demand meds used to treat breakthrough attacks at home.[5,6]
- Despite its effectiveness, additional in-depth research is needed to fully understand patients' experiences while using C1INH(SC).
- Qualitative patient interviews provide valuable insights into patient experiences, treatment satisfaction, adherence, and unmet needs. These insights can guide treatment optimization, enhance patient-centered care, and inform future research in the management of HAE.
- **This study aimed to gain insight into QOL impacts and treatment experience of routine LTP with C1INH(SC), along with on-demand medication use and breakthrough attacks, in patients with HAE via qualitative interviews.**

METHODS

STUDY DESIGN & SAMPLE SELECTION

- This study conducted semi-structured, qualitative interviews with adult participants (≥18 years) with HAE (Type I or Type II) who had been using LTP with C1INH(SC) for at least 1 year, and also using on-demand medication only for ≥1 year before starting C1INH(SC).
- Patient identification and recruitment: participants were selected from the practice populations of 7 highly experienced US clinician-investigators in HAE treatment.
- Note: the qualitative interviews were conducted as part of a larger hybrid retrospective study, in combination with a review of medical chart review data. For more information, see HAEA Poster #25.

DATA COLLECTION

- Patient interviews, lasting approximately 30 minutes in length each, were conducted via telephone by interviewers with expertise in qualitative interview methodology from ICON plc, using a semi-structured interview guide with open-ended prompts.
- Interviews were transcribed from audio recordings and de-identified. Transcripts were analyzed using cross-sectional thematic methodology with inductive and deductive approaches and coded using MAXQDA software to identify themes and information relating to study objectives.
- Patients received a $100 electronic gift card each as compensation.

RESULTS

STUDY SAMPLE

- The sample included **36 patients** aged 24–77 years (mean, 47.9) — see **Table 1**. Majority (94%) were HAE Type I.

TABLE 1. SOCIODEMOGRAPHIC AND CLINICAL CHARACTERISTICS OF STUDY SAMPLE

Characteristic	N=36
Age at Interview: Mean/Range (Standard Dev)	47.86 (±15.61), 24-77 years
Sex: Female	23 (64%)
Race/Ethnicity	
White	32 (89%)
Black or African American	3 (8%)
Other[a]	1 (3%)
Working Status	
Working full-time	22 (61%)
Retired	6 (17%)
Looking after home / family or unemployed / seeking work	2 (6%)
Self-employed	2 (6%)
Permanently unable to work / disability	2 (6%)
Working part-time	2 (6%)
Insurance[b]	
Private health insurance	25 (69%)
Medicare/Medicaid	11 (31%)
Other	5 (17%)
Patient Medical History	
HAE Type 1	34 (94%)
HAE Type 2	2 (6%)
Family history of HAE	29 (81%) (n=34)

Data are n (%) unless noted otherwise. N=36 unless noted otherwise. [a]"Other" reported as multiracial (White/Native Hawaiian/Hispanic/Latino); [b]Some patients reported multiple forms of insurance. HAE, hereditary angioedema; SD, standard deviation

STARTING TREATMENT WITH C1INH(SC)

- Reasons for starting C1INH(SC): ease of administration (42%), discussions with clinician (42%), challenges with previous meds (22%), impacts on children (17%), and frequent attacks (14%).
- Dosing frequencies: biweekly (67%) or every 3-4 days (31%). Ten reported changing frequency/dosage of C1INH(SC) (28%) citing a process of finding the best regimen, weight changes, and pregnancies.

FIGURE 1. IMPROVEMENTS TO QUALITY OF LIFE DOMAINS WHILE USING C1INH(SC)

Data represent the sample of patients whose interview content included mention of improved aspects of life **in the QOL domains**, when compared to their QOL before using C1INH(SC)

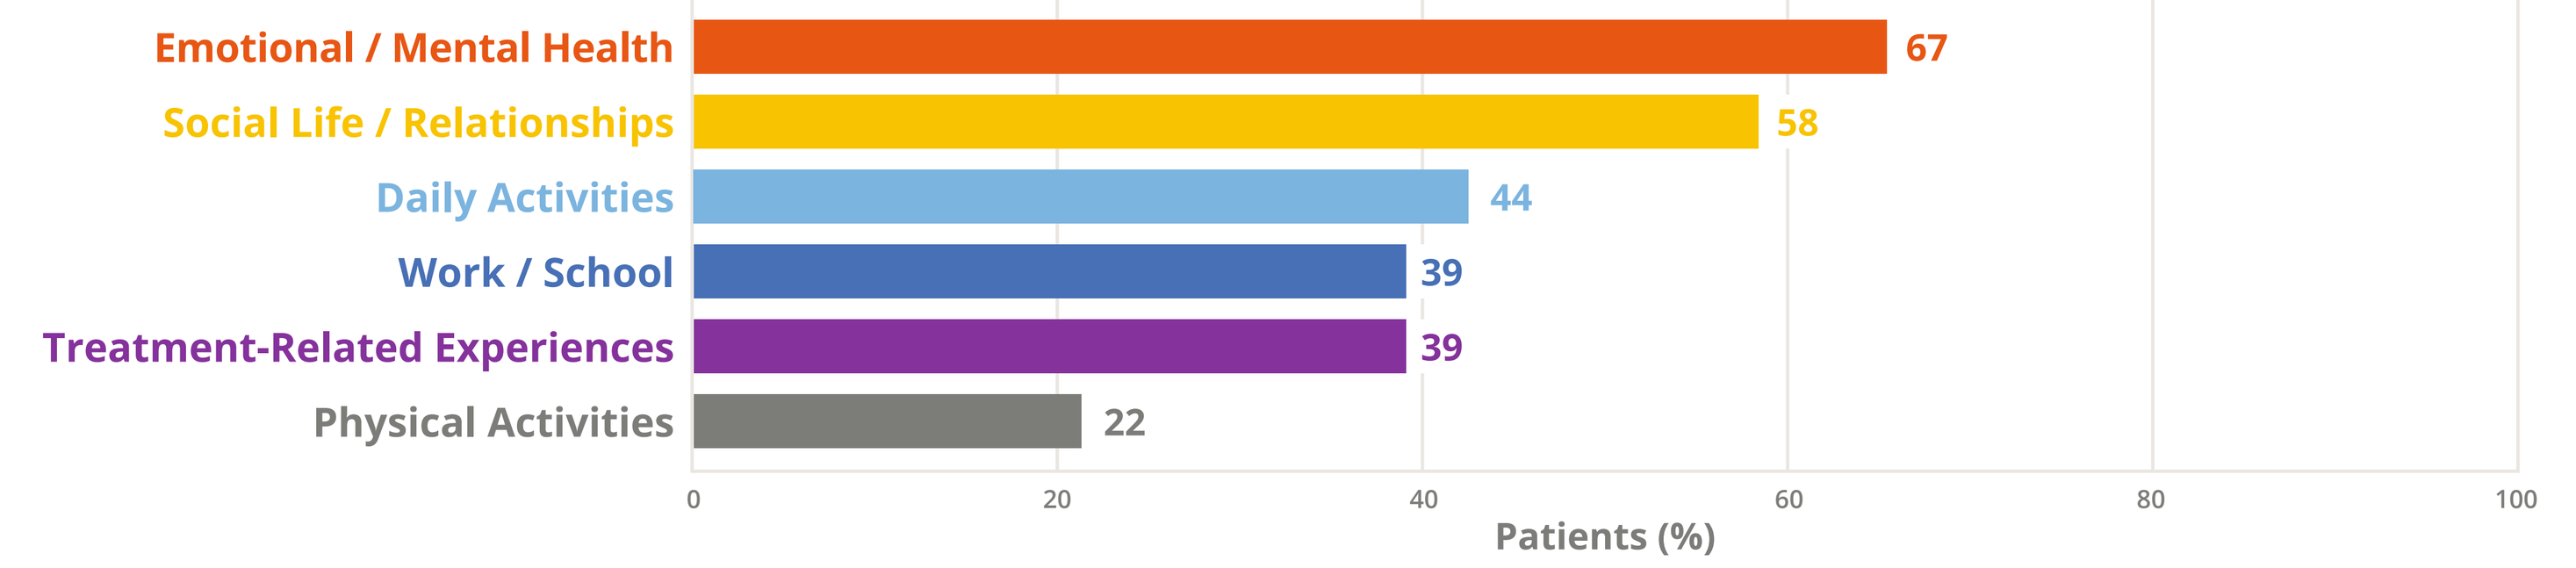
Emotional / Mental Health 67
Social Life / Relationships 58
Daily Activities 44
Work / School 39
Treatment-Related Experiences 39
Physical Activities 22
0
20
40
60
80
100
Patients (%)

HAE SYMPTOMOLOGY & BREAKTHROUGH ATTACKS

- Attack frequency:
 - **Before C1INH(SC):** between 1-5 per month (61%) on average, up to 15–18 per month or near-constant attacks.
 - **After starting C1INH(SC):** 94% reported a reduction in frequency — 0 or <1 per year (56%); none at all (33%).
- Attack severity:
 - **Before C1INH(SC):** from "mild" to "very severe," requiring hospitalization for many (67%) at its worst.
 - **After starting C1INH(SC):** 89% reported a reduction in severity — 3 reported no change (8%) but quicker resolution, reduced frequency. Only 2 reports of hospitalization (6%).
- Commonly reported symptoms:
 - **Before C1INH(SC):** swelling (94%), pain (61%), vomiting (56%), rash (31%), diarrhea (22%), and nausea (19%). All reported their **"worst attacks,"** (100%) commonly involving tracheotomy, intubation, difficulty breathing, and abdominal attacks.
 - **After starting C1INH(SC):** Decreased reports of symptoms post-C1INH(SC), while similar symptomology remained. Only 7 noted their "**worst attacks**" (19%).

QUALITY OF LIFE CHANGES

- **Before C1INH(SC):** QOL was negatively impacted pre-C1INH(SC) — including: emotional/mental health (89%), social life & relationships (86%), work/school (83%), physical activities (75%), daily activities (75%), and treatment experience (17%).
- **After starting C1INH(SC):** Improvements to QOL reported, including all aforementioned domains. See **Figure 1**.

BREAKTHROUGH ATTACK MANAGEMENT

- Management of attacks: Using on-demand meds (44%); pushing through or "waiting it out" (36%), going to the hospital (33%), resting or staying in bed (17%), and using pain medication (14%).
- After starting C1INH(SC), patients experiencing rare attacks managed with on-demand meds only.
- Untreated attacks: Pre-C1INH(SC): lasted from 1-2 days up to 2-3 weeks. Post-C1INH(SC): few hours up to 8 days.
- Treated with on-demand meds: resolved in few hours to 1-2 days for both instances; however, attack frequency and severity were significantly reduced after starting C1INH(SC).

ON-DEMAND MEDICATION USAGE

- On C1INH(SC) — reduced on-demand med use (92%); most required 1 (44%) or 2 doses (14%) to resolve.
- In this sample, 44% no longer used on-demand meds citing zero or very mild attacks on C1INH(SC).
- Patients generally kept multiple doses of on-demand meds, for short-term use (47%), due to insurance challenges (25%) and concerns about availability (17%) Most requested refills themselves (69%).
- Patients shared medication with family members (n=12, 33%) and used/retained expired meds (11%).

TABLE 2. ILLUSTRATIVE QUOTES FROM PATIENT INTERVIEWS BY TOPIC

Starting Treatment with C1INH(SC)

PT-D-002: My doctor had told me that there was a new sub-Q injection that [I] could do that [they] had good results with ... Takhzyro I had to stop because I was having an allergic reaction ... and it wasn't working for me.

PT-C-003: So what I liked about [C1INH(SC)] .. it's so much easier to administer than an IV, because it is sub-Q.

PT-C-003: Personally, I prefer one that is a C1 replacement therapy, and had such a great experience on the trial that I didn't want to go back to living without it.

Breakthrough Attacks: Changes in Frequency and Severity

PT-E-003: I have had no swelling. No attacks whatsoever since taking [C1INH(SC)]. So the [C1INH(SC)] has been a really lifesaving experience for me.

PT-G-009: Yes. The frequency went down quite a bit. So I was very, very grateful for that.

PT-F-006: It's definitely less severe since being on [C1INH(SC)] ... I very rarely get a hand or a foot swell at all.

PT-G-003: I haven't had any attacks since I started the study. It's why I love talking about [C1INH(SC)].

PT-C-005: Since [C1INH(SC)] I would say it was maybe one a year, and ... I haven't had an attack in like a few years.

Quality of LIfe Changes

PT-A-001: It does provide more stability in my life, so it makes it a lot easier to be consistently working, and makes it easier to make commitments to things, socially. Eliminate[s] some of the burden of the disorder.

PT-G-009: I felt a lot better about my life in general because I had more time to do the stuff that I need to do. I was in the hospital less, which was wonderful.

PT-D-004: Yes. All to the positive. Because with the reduction of attacks ... you've got some control over the disease and a regimen that's working ... participating in more family events, a lot fewer doctor visits and hospital visits. The sense of security.

PT-D-001: It's amazing what these drugs have done. Emotionally, physically, and especially with [C1INH(SC)] being sub[-Q].

PT-G-011: It's just a great blessing now that I don't have any [attacks].

PT-E-001: It just changes everything when it's not in the back of your mind constantly. You know that you are going to have a good day every day ... It just makes a huge difference in your life.

Breakthrough Attacks & On-Demand Medications

PT-E-001: Typically what I would do is wait [the attack] out a little bit to see if it's going to pass.

PT-H-001: When something is starting to swell, I will just go ahead and use Firazyr... Why mess with it if it works?

PT-D-001: Untreated anywhere from 3-5 days. A treated attack, it varied, but 24 hours ... I could resume normal activities.

PT-B-001: When the medication expires, we toss it out. The Firazyr is a syringe — so we have a sharps container ... The Berinert is just a vial, so that just gets tossed out.

PT-G-011: I do have a couple of doses on hand just in case there would be a breakthrough attack ever.

PT-C-003: Yes [to sharing]. My daughter has a prescription for the same thing and there have been times when she needed it and she was between treatments.

DISCLOSURES

Funding Source: Research and medical writing support funded and provided by CSL Behring to ICON plc.

REFERENCES

1. Bork K, Anderson JT, Caballero T, et al. Allergy Asthma Clin Immunol. 2021;17:40. **2.** Caballero T, Aygoren-Pursun E, Bygum A, et al. Allergy Asthma Proc. 2013;34:1-7. **3.** Lumry WR, Craig T, Zuraw B, et al. J Allergy Clin Immunol Pract. 2018;6(5):1733-1741. **4.** Lumry WR, Zuraw B, Cicardi M, et al. Orphanet J Rare Dis. 2021;16:86. **5.** Maurer M, Magerl M, Betschel S, et al. Allergy. 2022;77:1961-1990. **6.** Busse PJ, Christiansen SC, Riedl MA, et al. J Allergy Clin Immunol Pract. 2021;9:132-150.

Presented at the 2023 US HAEA National Summit, Orlando, FL, July 20-23, 2023.