Garadacimab For Hereditary Angioedema Prophylaxis In Adolescents: Efficacy And Safety From The Phase 3 (VANGUARD) Study And Open-Label Extension (Second Interim Analysis)

P016

Joshua S Jacobs,[1] Markus Magerl,[2,3] Inmaculada Martinez-Saguer,[4] H Henry Li,[5] Jonathan A Bernstein,[6] Connie Hsu,[7] Karl Sitz,[8] Henrike Feuersenger,[9] Lolis Wieman,[10] Maressa Pollen,[10] Avner Reshef[11]

[1]Allergy & Asthma Clinical Research, Walnut Creek, CA, USA; [2]Institute of Allergology, Charité–Universitätsmedizin Berlin, Berlin, Germany; [3]Fraunhofer Institute for Translational Medicine and Pharmacology ITMP, Immunology and Allergology, Berlin, Germany; [4]HZRM Haemophilia Centre Rhein Main, Mörfelden-Walldorf, Germany; [5]Institute for Asthma and Allergy, Chevy Chase, MD, USA; [6]University of Cincinnati, Department of Internal Medicine Division of Rheumatology, Allergy and Immunology and the Bernstein Clinical Research Center Cincinnati, Cincinnati, OH, USA; [7]Research Solutions of Arizona, PC, Litchfield Park, AZ, USA; [8]Little Rock Allergy and Asthma Clinic, Little Rock, AR, USA; [9]CSL Behring Innovation GmbH, Marburg, Germany; [10]CSL Behring, King of Prussia, PA, USA; [11]Allergy, Immunology & Angioedema Center, Barzilai University Hospital, Ashkelon, Israel.

Conclusions

- **Garadacimab provided durable efficacy and sustained protection against hereditary angioedema (HAE) attacks in adolescents in the Phase 3 open-label extension (OLE) study, consistent with the pivotal Phase 3 study**
- **Garadacimab demonstrated a favorable long-term safety profile in adolescents (aged 12 to ≤17 years) with HAE in the pivotal Phase 3 (VANGUARD)[1] and ongoing Phase 3 OLE studies[2]**
- **A Phase 3 study evaluating garadacimab in pediatric patients (aged 2–11 years) with HAE is ongoing; five sites are activated in the USA, Canada, and Israel, with more sites to be activated in future (NCT05819775, EudraCT: 2022-502386-13-00)[3]**

BACKGROUND

HAE[4–6]

- HAE causes recurrent, unpredictable, debilitating, and potentially life-threatening attacks
- HAE symptoms typically begin in the first two decades of life, with attack intensity worsening during puberty
- Dysregulation of the kallikrein–kinin pathway leads to uncontrolled production of bradykinin, the key mediator of HAE attacks
- Activated factor XII (FXIIa) initiates the kallikrein–kinin pathway

Garadacimab[1,7]

- First-in-class, fully human, monoclonal antibody targeting FXIIa for the prophylaxis of HAE attacks
- High affinity, potency, and specificity for FXIIa, shown to decrease bradykinin production
- In the pivotal Phase 3 (VANGUARD) and Phase 3 OLE (NCT04739059) studies, garadacimab once-monthly demonstrated durable efficacy with a favorable long-term safety profile

OBJECTIVE

- To report long-term efficacy and safety in the adolescent subgroups from the pivotal Phase 3 (n=4) and ongoing Phase 3 OLE (n=10) studies (data cut-off: February 13, 2023; NCT04739059)

PIVOTAL PHASE 3 AND OLE STUDY DESIGN

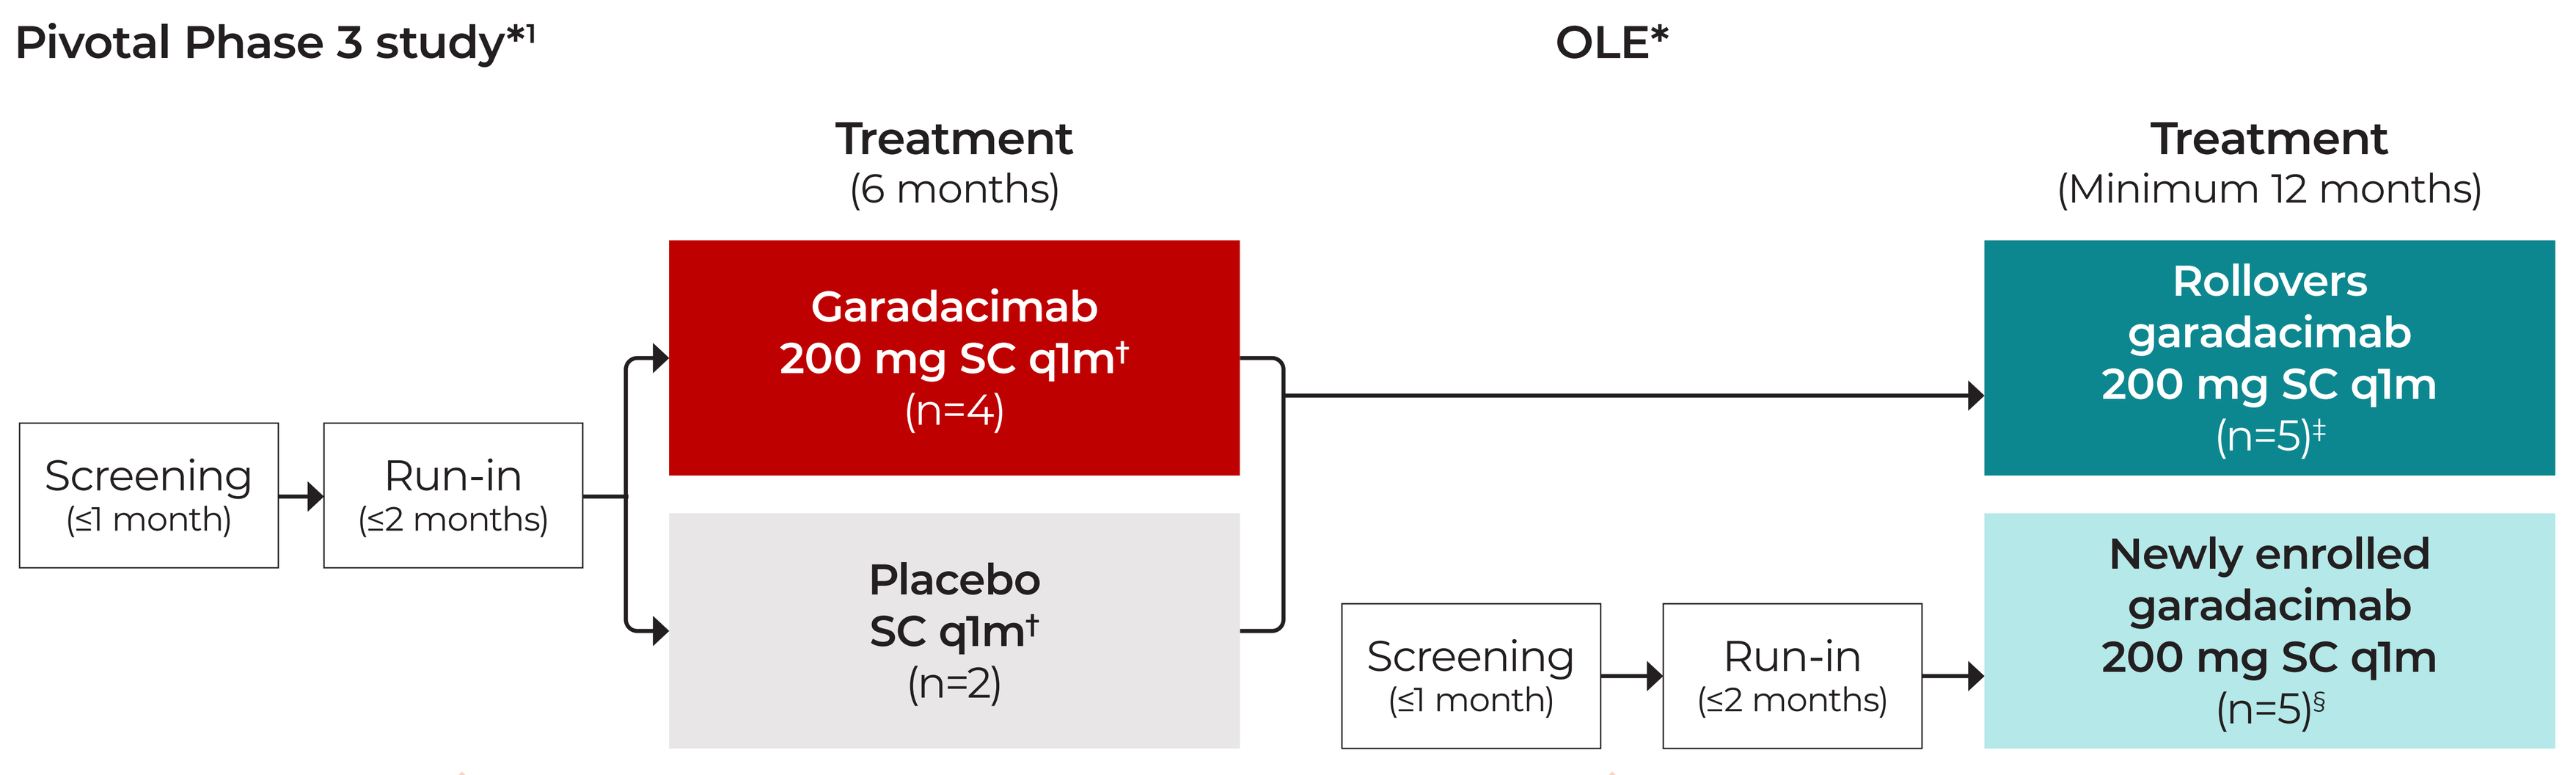
Pivotal Phase 3 study*1
OLE*
Screening (≤1 month)
Run-in (≤2 months)
Treatment (6 months)
Garadacimab 200 mg SC q1m† (n=4)
Placebo SC q1m† (n=2)
Screening (≤1 month)
Run-in (≤2 months)
Treatment (Minimum 12 months)
Rollovers garadacimab 200 mg SC q1m (n=5)‡
Newly enrolled garadacimab 200 mg SC q1m (n=5)§

Pivotal Phase 3 study endpoints
- Primary: Time-normalized number of attacks
- Key secondary: Reduction in attack rate vs run-in and safety

OLE study endpoints
- Primary: TEAEs
- Key secondary: Time-normalized number of attacks, and reduction in attack rate vs run-in

All patients were permitted to use on-demand therapy to treat HAE attacks throughout the pivotal Phase 3 and OLE studies.
*Eligibility criteria for entry into the pivotal Phase 3 and OLE studies included clinically confirmed HAE, age ≥12 years, and baseline attack rate ≥1 HAE attack/month; †In the pivotal Phase 3 study, patients received a loading dose of garadacimab 400 mg SC as first dose (two injections of 200 mg) or volume-matched placebo; ‡One patient aged 18 years at OLE enrollment was considered an adult; therefore, they were no longer included in the adolescent analysis; §Newly enrolled patients in the OLE study received a loading dose of garadacimab 400 mg SC as first dose (two injections of 200 mg).
HAE, hereditary angioedema; OLE, open-label extension; q1m, once-monthly (every 30 ± 4 days); SC, subcutaneous; TEAE, treatment-emergent adverse event.

RESULTS

Garadacimab reduced mean monthly attack rate in adolescents

- Garadacimab reduced mean monthly attack rate by 91% (95% CI 82.9–100) vs run-in in adolescents in the OLE study (0.09 vs 1.86 at run-in)

Patient*	Sex	Garadacimab exposure (months)† Median exposure 16.3 months	Monthly attack rate (% reduction vs run-in): Run-in	Pivotal Phase 3 study	OLE study Median exposure 13.8 months
1	F	16.7	1.2	1.2 (−5.8)	0.4 (68.2)
2	M	17.5	2.2	0.2 (92.3)	0.1 (96.0)
3	F	6.0†	2.9	**0** (100)‡	§
4	M	16.3	1.2	**0** (100)‡	0.1 (92.0)
5	M	12.8	1.0	Placebo: 1.2 (−12.2)	0.1 (92.4)
6	F	13.8	1.8	Placebo: 0.2 (90.8)	**0** (100)‡
7	M	7.7	0.9	–	0.3 (71.1)
8	F	19.7	1.0	–	0.1 (95.2)
9	F	17.0	4.9	–	**0** (100)‡
10	F	8.9	3.2	–	**0** (100)‡
11	M	8.0	1.2	–	**0** (100)‡

*All adolescents (aged 12 to ≤17 years) had HAE type 1, were White, and had a BMI range of 17.9–42.3 kg/m^2; †Garadacimab exposure inclusive of the pivotal Phase 3 and OLE studies as appropriate; as Patient 3 was aged 18 years at OLE enrollment, they were considered an adult and only exposure in pivotal Phase 3 study is reported; ‡Attack-free patients; §Patient aged 18 years at OLE enrollment was considered an adult; therefore, they were no longer included in the adolescent analysis.
BMI, body mass index; CI, confidence interval; F, female; HAE, hereditary angioedema; M, male; OLE, open-label extension.

Favorable long-term safety profile in adolescents

AEs	Pivotal Phase 3 study (Exposure: 6 months) Placebo (n=2)	Pivotal Phase 3 study (Exposure: 6 months) Garadacimab (n=4)	OLE study (Median exposure: 13.8 months) Garadacimab (n=10)*
Any TEAEs, n (%)	2 (100)	2 (50)	7 (70)
Related to treatment, n	0	0	0
Death, n	0	0	0
TEAEs leading to discontinuation, n	0	0	0
TEAEs by severity			
Mild, n (%)	2 (100)	1 (25)	7 (70)
Moderate,† n (%)	0	1 (25)	2 (20)
Severe, n	0	0	0
SAEs, n	0	0	0
AESIs per protocol,‡ n	0	0	0
Injection-site reactions, n	0	0	0
Most common TEAEs in ≥2 adolescents in the OLE study			
COVID-19, n (%)	2 (100)	0	3 (30)
Nasopharyngitis, n (%)	0	1 (25)	3 (30)

*One patient aged 18 years at OLE enrollment was considered an adult; therefore, they were no longer included in the adolescent analysis; †Includes sore throat (pivotal Phase 3 study); upper respiratory tract infection (two occurrences), flu-like symptoms, COVID-19, and acid reflux (OLE); ‡Includes abnormal bleeding events, thromboembolic events, and severe hypersensitivity, including anaphylaxis.
AE, adverse event; AESI, adverse event of special interest; COVID-19, coronavirus disease 2019; OLE, open-label extension; SAE, serious adverse event; TEAE, treatment-emergent adverse event.

Garadacimab provides early onset of protection against HAE attacks with durable efficacy in adolescents

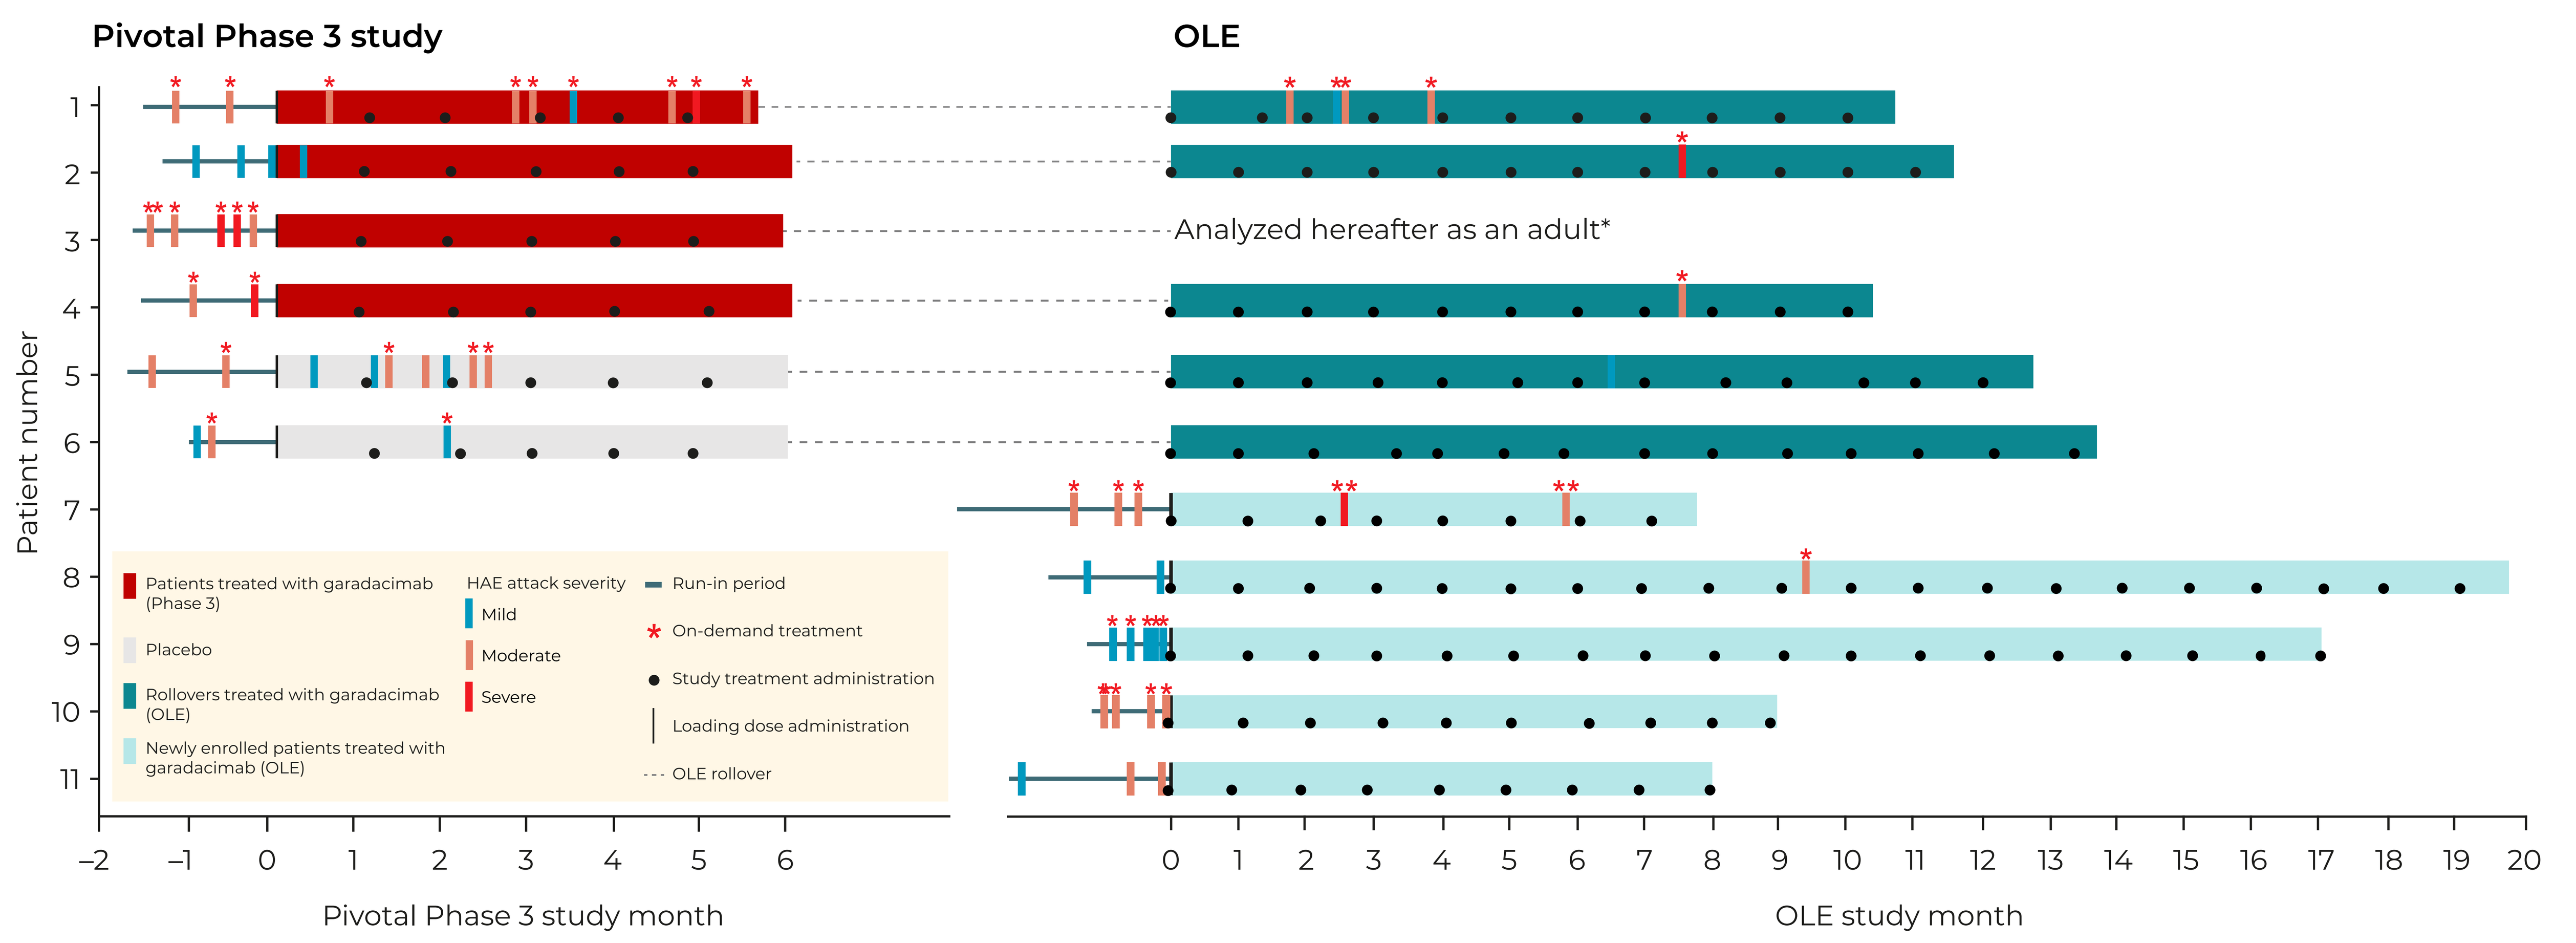
Pivotal Phase 3 study
OLE
Patient number
Analyzed hereafter as an adult*
Patients treated with garadacimab (Phase 3)
Placebo
Rollovers treated with garadacimab (OLE)
Newly enrolled patients treated with garadacimab (OLE)
HAE attack severity
Mild
Moderate
Severe
Run-in period
On-demand treatment
Study treatment administration
Loading dose administration
OLE rollover
Pivotal Phase 3 study month
OLE study month

*Patient aged 18 years at OLE enrollment; therefore, they were considered an adult.
OLE, open-label extension.

Pivotal Phase 3 study entrants (Patients 1–6)
(range of exposure: 6.0–17.5 months)

Received garadacimab in the pivotal Phase 3 study (Patients 1–4):
- **Pivotal:** ≥90% attack rate reductions vs run-in: 3/4 (75%) adolescents
 - **2**/4 (50%) were attack free
- **OLE:** ≥68% attack rate reductions vs run-in in three rollover adolescents

Received placebo in the pivotal Phase 3 study (Patients 5 and 6):
- **OLE:** 92.4% attack rate reduction vs run-in in one adolescent, the other was attack free

Newly enrolled patients (Patients 7–11)
(range of exposure: 7.7–19.7 months)

- ≥70% attack rate reductions vs run-in in all five newly enrolled adolescents
 - 3/5 (60%) were attack free in the entire OLE study

High responder rates were observed in adolescents in the OLE study

All patients receiving garadacimab experienced **≥50% reduction** in attack rate vs run-in

80% of patients experienced **≥90% reduction** in attack rate vs run-in

Scan the QR code to access the poster.

References

1. Craig TJ et al. *Lancet* 2023;401:1079–1090; 2. ClinicalTrials.gov. NCT04739059. Available at: https://classic.clinicaltrials.gov/ct2/show/NCT04739059 (accessed January 2024); 3. ClinicalTrials.gov. NCT05819775. Available at: https://clinicaltrials.gov/study/NCT05819775 (accessed January 2024); 4. Busse PJ et al. *J Allergy Clin Immunol Pract* 2021;9:132–150.e3; 5. Davis AE 3rd. *Transfus Apher Sci* 2003;29:195–203; 6. Bygum A et al. *Clin Transl Allergy* 2019;9:37; 7. Cao H et al. *J Allergy Clin Immunol* 2018;142:1355–1358.

Disclosures

Writing support provided by OPEN Health Scientific Communications funded by CSL Behring.

American Academy of Allergy, Asthma, and Immunology Annual Meeting | Washington DC, USA | February 23–26, 2024