Garadacimab Pharmacokinetic and Safety Profiles Show No Impact of Concomitant Medication Use in Hereditary Angioedema

628

Paul K Keith[1], John Anderson[2], H Henry Li[3], Markus Magerl[4–6], Regina Treudler[4], Fiona Glassman[7*], Harsha Shetty[8], John-Philip Lawo[9], Maressa Pollen[8], Timothy J Craig[10,11]

[1]McMaster University Medical Centre, Hamilton, ON, Canada; [2]AllerVie Clinical Research, Birmingham, AL, USA; [3]Institute for Asthma and Allergy, Chevy Chase, MD, USA; [4]Institute of Allergology, Charité–Universitätsmedizin Berlin, Berlin, Germany; [5]Freie Universität Berlin and Humboldt-Universität zu Berlin, Berlin, Germany; [6]Fraunhofer Institute for Translational Medicine and Pharmacology (ITMP), Immunology and Allergology, Berlin, Germany; [7]Biogen, Cambridge, MA, USA; [8]CSL Behring, King of Prussia, PA, USA; [9]CSL Innovation GmbH, Marburg, Germany; [10]Allergy, Asthma and Immunology, Department of Medicine and Pediatrics, Pennsylvania State University, Hershey, PA, USA; [11]Vinmec International Hospital, Times City, Hanoi, Vietnam. *After completion of the study, Fiona Glassman moved from CSL Behring, King of Prussia, PA, USA, to Biogen, Cambridge, MA, USA.

CONCLUSIONS

- **No safety concerns were identified for patients receiving garadacimab and concomitant analgesic, antihistamine, anti-inflammatory, or antibacterial medications**
- **Garadacimab had no impact on hemostasis in patients receiving concomitant hemostasis-impacting medication, and its safety profile was consistent with that of patients receiving garadacimab alone**
- **The pharmacokinetic (PK) profile of garadacimab was unaffected by the use of concomitant medications**

BACKGROUND

- Hereditary angioedema (HAE) causes recurrent, unpredictable, debilitating, and potentially life-threatening attacks[1–5]
- Garadacimab is a first-in-class, fully human, anti-activated factor XII monoclonal antibody under evaluation for the long-term prophylaxis of HAE attacks[1,2,6]
- Garadacimab has demonstrated early and durable efficacy with a favorable long-term safety profile in Phase 2, pivotal Phase 3 (VANGUARD) and Phase 3 open-label extension (OLE) studies[1,2,7]
 - Garadacimab had no impact on hemostasis in these studies[1,2,7,8]
- Pharmacokinetic (PK) modelling of garadacimab was unaffected by covariates including age, sex, race, HAE subtype, use of concomitant medication or on-demand therapy for HAE, and immunogenicity[9]

OBJECTIVE

- To evaluate the impact of concomitant medications commonly used in clinical practice on the PK and safety profiles of garadacimab

STUDY DESIGN: SAFETY AND PK ANALYSES

- *Post hoc* review of treatment-emergent adverse events (TEAEs) coded by Medical Dictionary for Regulatory Activities terminology was performed for patients who received garadacimab and concomitant analgesic, antihistamine, anti-inflammatory, or antibacterial medications* in the Phase 2, pivotal Phase 3, and Phase 3 OLE studies
- Adverse events of special interest (AESIs) per protocol (abnormal bleeding, thromboembolic events, severe hypersensitivity, and anaphylaxis) monitored in the Phase 2, pivotal Phase 3 (VANGUARD), and ongoing Phase 3 OLE studies[1,2,7] underwent medical review for patients receiving garadacimab and concomitant hemostasis-impacting medications†
- A population PK model was built using data pooled from five clinical studies (Phase 1–3) to assess PK for garadacimab
- Predicted PK exposure metrics for patients who received concomitant medication were compared with those who did not, using empirical Bayes estimates

*Analgesics included acetaminophen, non-steroidal anti-inflammatory drugs, amitriptyline, cannabidiol, cyclo-oxygenase inhibitors, gabapentin, galcanezumab, metamizole, opiates, pheniramine, pregabalin, scutellaria, sumatriptan, sympathomimetics, topiramate, and ubrogepant; antihistamines included bilastine, cetirizine, chlorpheniramine, clemastine, desloratadine, diphenhydramine, ebastine, epinastine, famotidine, fexofenadine, hydroxyzine, levocetirizine, loratadine, moxastine, olopatadine, promethazine, pseudoephedrine, and rupatadine; anti-inflammatories included acetaminophen, non-steroidal anti-inflammatory drugs, icatibant, glucosamine and chondroitin, pseudoephedrine, and sulfasalazine; antibacterials included amoxicillin, azithromycin, cephalosporins, D-mannose, and metronidazole; †Factor Xa inhibitors, direct thrombin inhibitors, anticoagulants, and heparins were classed as medications with direct impact on hemostasis; medications for thrombocytopenia or for platelet dysfunction were classed as medications with indirect impact on hemostasis.

PK model data pooled from five clinical studies[8]

Healthy adults (aged >17 years)

- **Phase 1 randomized, placebo-controlled, single-ascending dose study (n=48)[10]**: 0.1–10 mg/kg IV or 1–10 mg/kg SC[10]
- **Phase 1 ethnobridging, single-ascending dose study (n=37)[11]***: 3 or 10 mg/kg IV or 200 or 600 mg SC[11]

Adult patients with HAE (aged >17 years)

- **Phase 2 study (n=44)[6,7]**: IV loading dose† and 75, 200, or 600 mg SC q1m

Adult and adolescent patients with HAE (aged ≥12 years)

- **Pivotal Phase 3 study (n=64)[1]**: Loading dose (2 × 200 mg SC injections) 200 mg SC q1m
- **Phase 3 OLE (n=161)[2]**: Loading dose (2 × 200 mg SC injections) 200 mg SC q1m

N=262 unique participants with HAE and evaluable concentrations of garadacimab

*Healthy White adults were matched for body weight (±15%) 1:1 with healthy Japanese adults; †IV loading doses of placebo or garadacimab 40, 100, or 300 mg, followed by placebo or garadacimab 75, 200, or 600 mg SC, respectively.
HAE, hereditary angioedema; IV, intravenous; OLE, open-label extension; PK, pharmacokinetics; q1m, once monthly; SC, subcutaneous.

REFERENCES:
1. Craig TJ et al. *Lancet* 2023;401:1079–1090; 2. Reshef A et al. *Allergy* 2025;80:545–556; 3. Busse PJ et al. *J Allergy Clin Immunol Pract* 2021;9:132–150.e3; 4. Davis AE 3rd. *Transfus Apher Sci* 2003;29:195–203; 5. Maurer M et al. *Allergy* 2022;77:1961–1990; 6. Craig TJ et al. *Lancet Hematol* 2024;11:e436–e447; 7. Craig TJ et al. *Lancet* 2022;399:945–955; 8. Cohn D et al. *Ann Allergy Asthma Immunol* 2024;S31-S32; 9. Glassman F et al. *J Allergy Clin Immunol* 2024;153:AB4; 10. McKenzie A et al. *Clin Transl Sci* 2022;15: 626–637; 11. Glassman F et al. *J Clin Pharmacol* 2024 doi: 10.1002/jcph.6162 (online ahead of print).

RESULTS

Table 1: No safety concerns were reported with garadacimab when administered alongside common classes of concomitant medications

All garadacimab-related TEAEs were **mild or moderate** in severity

Of the TEAEs reported, **most were mild or moderate** and **most resolved** by end of treatment, consistent with previously reported safety profile of garadacimab

In this analysis, the most commonly reported TEAEs related to garadacimab were **injection-site reactions**

TEAEs on treatment with garadacimab whilst receiving concomitant medication, n (%)	Class of concomitant medication: Analgesics (n=99)	Antihistamines (n=48)	Anti-inflammatories (n=97)	Antibacterials (n=61)
Any TEAEs	711 (100)	299 (100)	612 (100)	498 (100)
Related to study treatment	63 (8.9)	27 (9.0)	41 (6.6)	41 (8.2)
TEAEs by severity*				
Mild	443 (62.3)	171 (57.2)	349 (57.0)	289 (58.0)
Moderate	250 (35.2)	120 (40.1)	248 (40.5)	199 (40.0)
Severe	16 (2.3)	8 (2.7)	14 (2.3)	8 (1.6)
TEAEs by outcome				
Recovered/resolved	610 (85.8)	258 (79.6)	521 (85.1)	446 (89.5)
Not recovered/resolved	62 (8.7)	39 (13.0)	65 (10.6)	33 (6.6)
Recovering	30 (4.2)	19 (6.4)	20 (3.3)	13 (2.6)
Recovered with sequelae	6 (0.8)	2 (0.7)	5 (0.8)	5 (1.0)
Injection-site reactions	42 (5.9)	17 (5.7)	24 (3.9)	30 (6.0)
TEAEs by SOC, >10% of patients†				
Infections and infestations	203 (28.6)	87 (29.1)	169 (27.6)	164 (32.9)
Gastrointestinal	102 (14.3)	<10%	65 (10.6)	59 (11.8)
General disorders and administration site conditions	99 (13.9)	36 (12.0)	76 (12.4)	69 (13.8)
Musculoskeletal and connective tissue disorders	<10%	35 (11.7)	62 (10.1)	<10%

No drug interactions related to concomitant medications were reported in the garadacimab clinical program

*Note data on TEAE severity was unavailable for n=3 patients receiving analgesics and n=1 patients in each of the other groups; †Infections and infestations: COVID-19, nasopharyngitis, URTI, oral herpes; gastrointestinal: Abdominal pain, nausea, toothache; General disorders and administration site conditions: Injection-site reactions (including pain, erythema, and urticaria), chest discomfort; Musculoskeletal: Arthralgia.
SOC, system organ class; TEAE, treatment-emergent adverse event, URTI, upper respiratory tract infection.

Concomitant hemostasis-affecting medications had no impact on the safety profile nor PK of garadacimab

Concomitant **hemostasis-impacting medications** did not impact the safety profile of garadacimab, **no impact on hemostasis** was observed*

Concomitant administration of **hemostasis-impacting medication** did **not affect garadacimab PK**

Figure 1: Predicted PK parameters for garadacimab with concomitant medications having A) a direct or B) an indirect impact on hemostasis

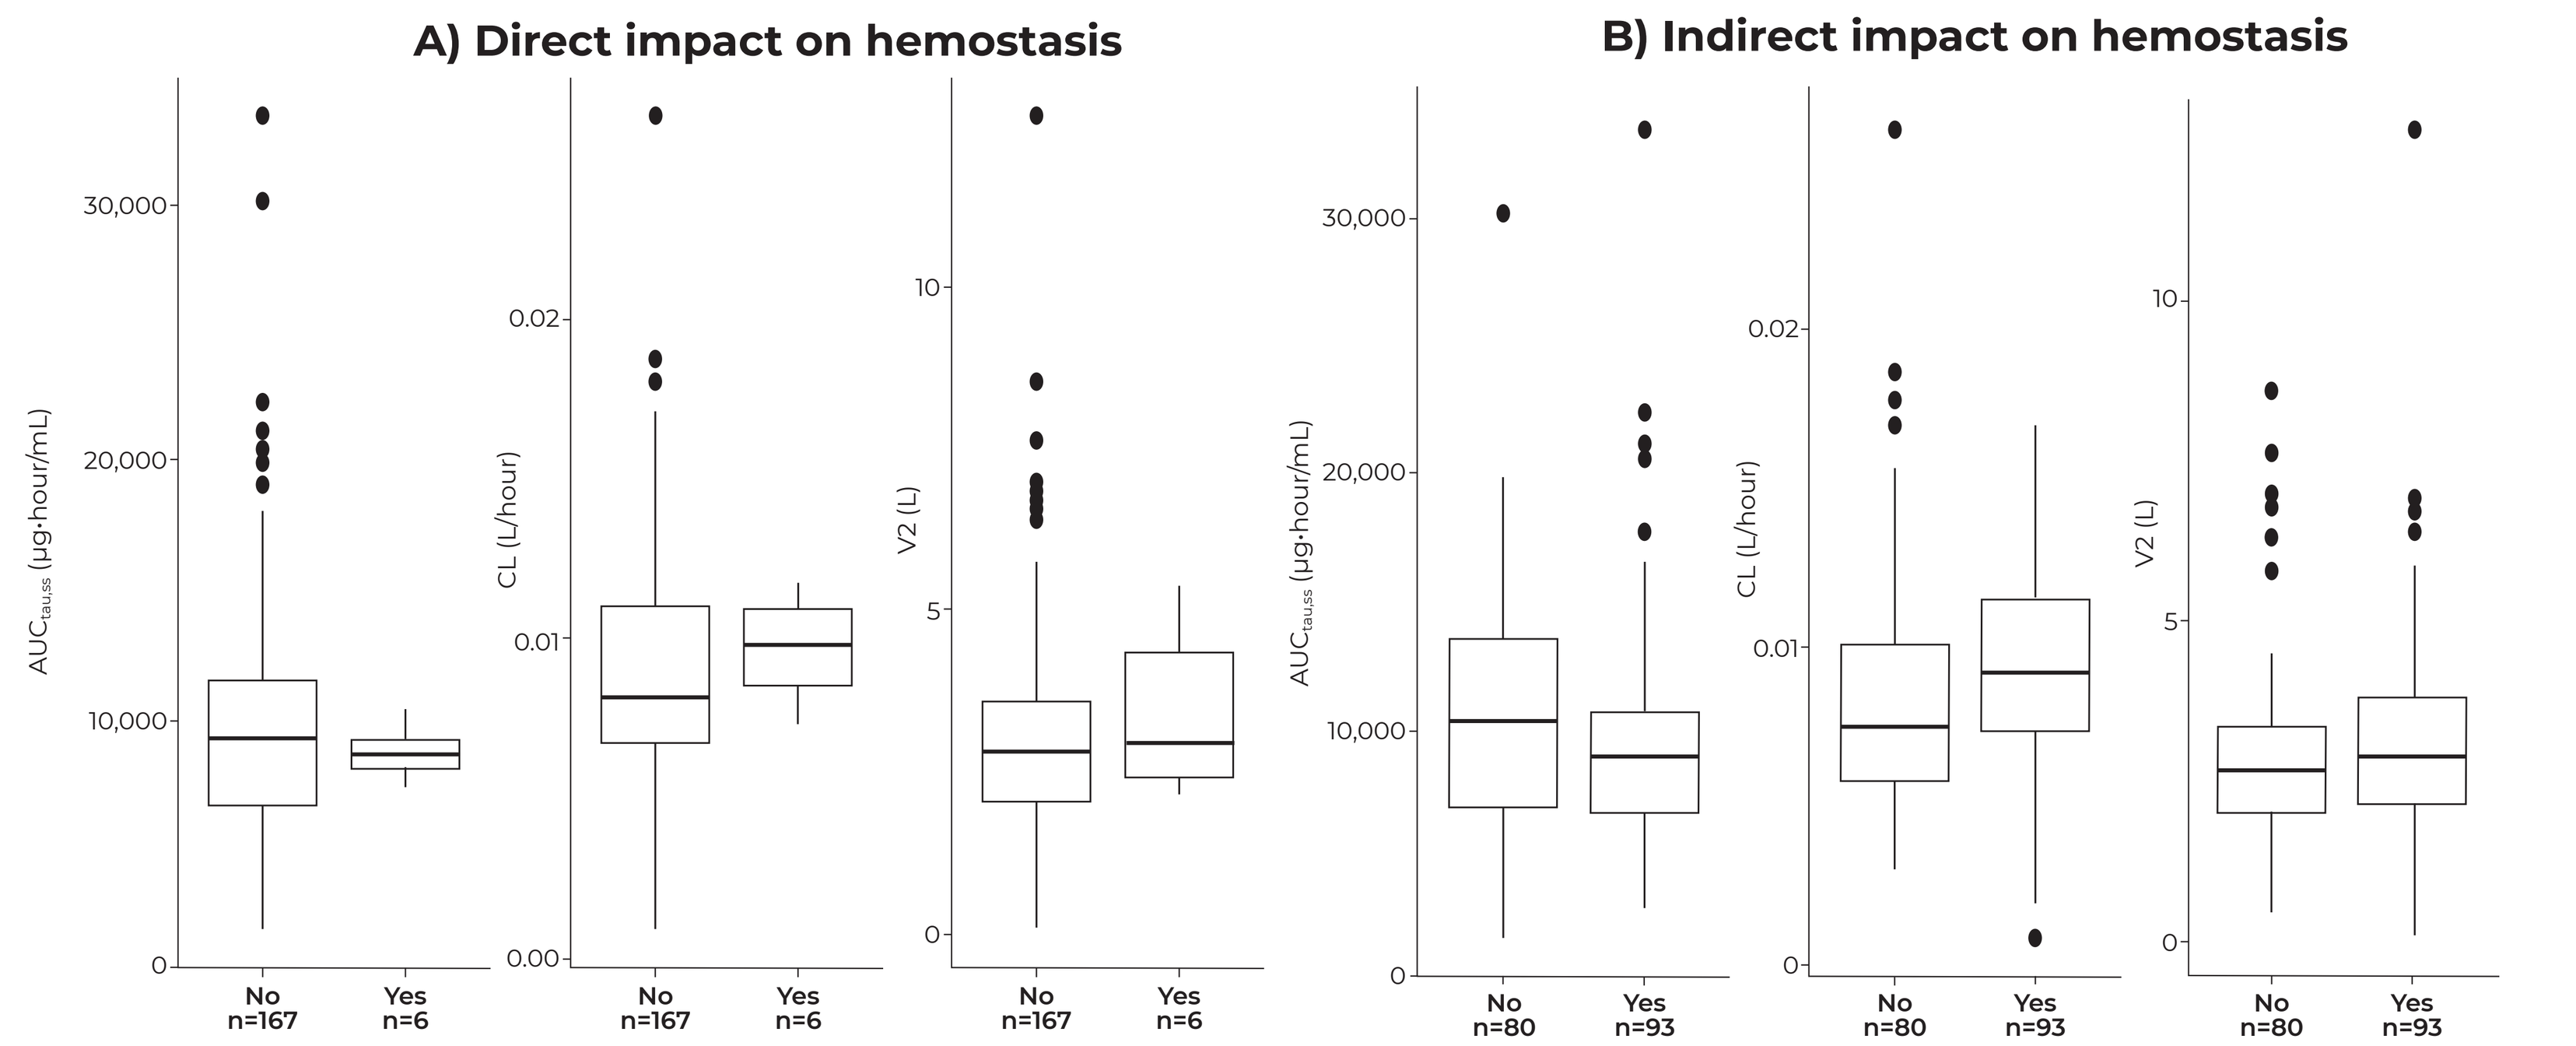
A) Direct impact on hemostasis
B) Indirect impact on hemostasis
AUC$_{tau,ss}$ (µg·hour/mL)
CL (L/hour)
V2 (L)
No n=167
Yes n=6
No n=80
Yes n=93

In the box plots, median values are represented by a solid line in the center of the box. Boxes indicate the IQR, with whiskers extending to 1.5 × IQR.
*Investigators reported bleeding events in one patient in the Phase 2 open-label period, 2 in the Phase 3 OLE; none were related to garadacimab and the event identified during the Phase 2 open-label period mild epistaxis was identified as an AESI per protocol in a patient receiving 600 mg garadacimab in the Phase 2 HAE study, which resolved by the end of the study.
AESI, adverse event of special interest; AUC$_{tau,ss}$, area under the concentration–time curve for a dosing interval at steady state; CL, total drug clearance; HAE, hereditary angioedema; IQR, interquartile range; OLE, open-label extension; PK, pharmacokinetics; V2, volume of distribution.

No impact of common concomitant medications on the PK of garadacimab

Figure 2: Predicted PK parameters for garadacimab in the presence of or without concomitant medication with: A) analgesics, B) antihistamines, C) anti-inflammatories, and D) antibacterials

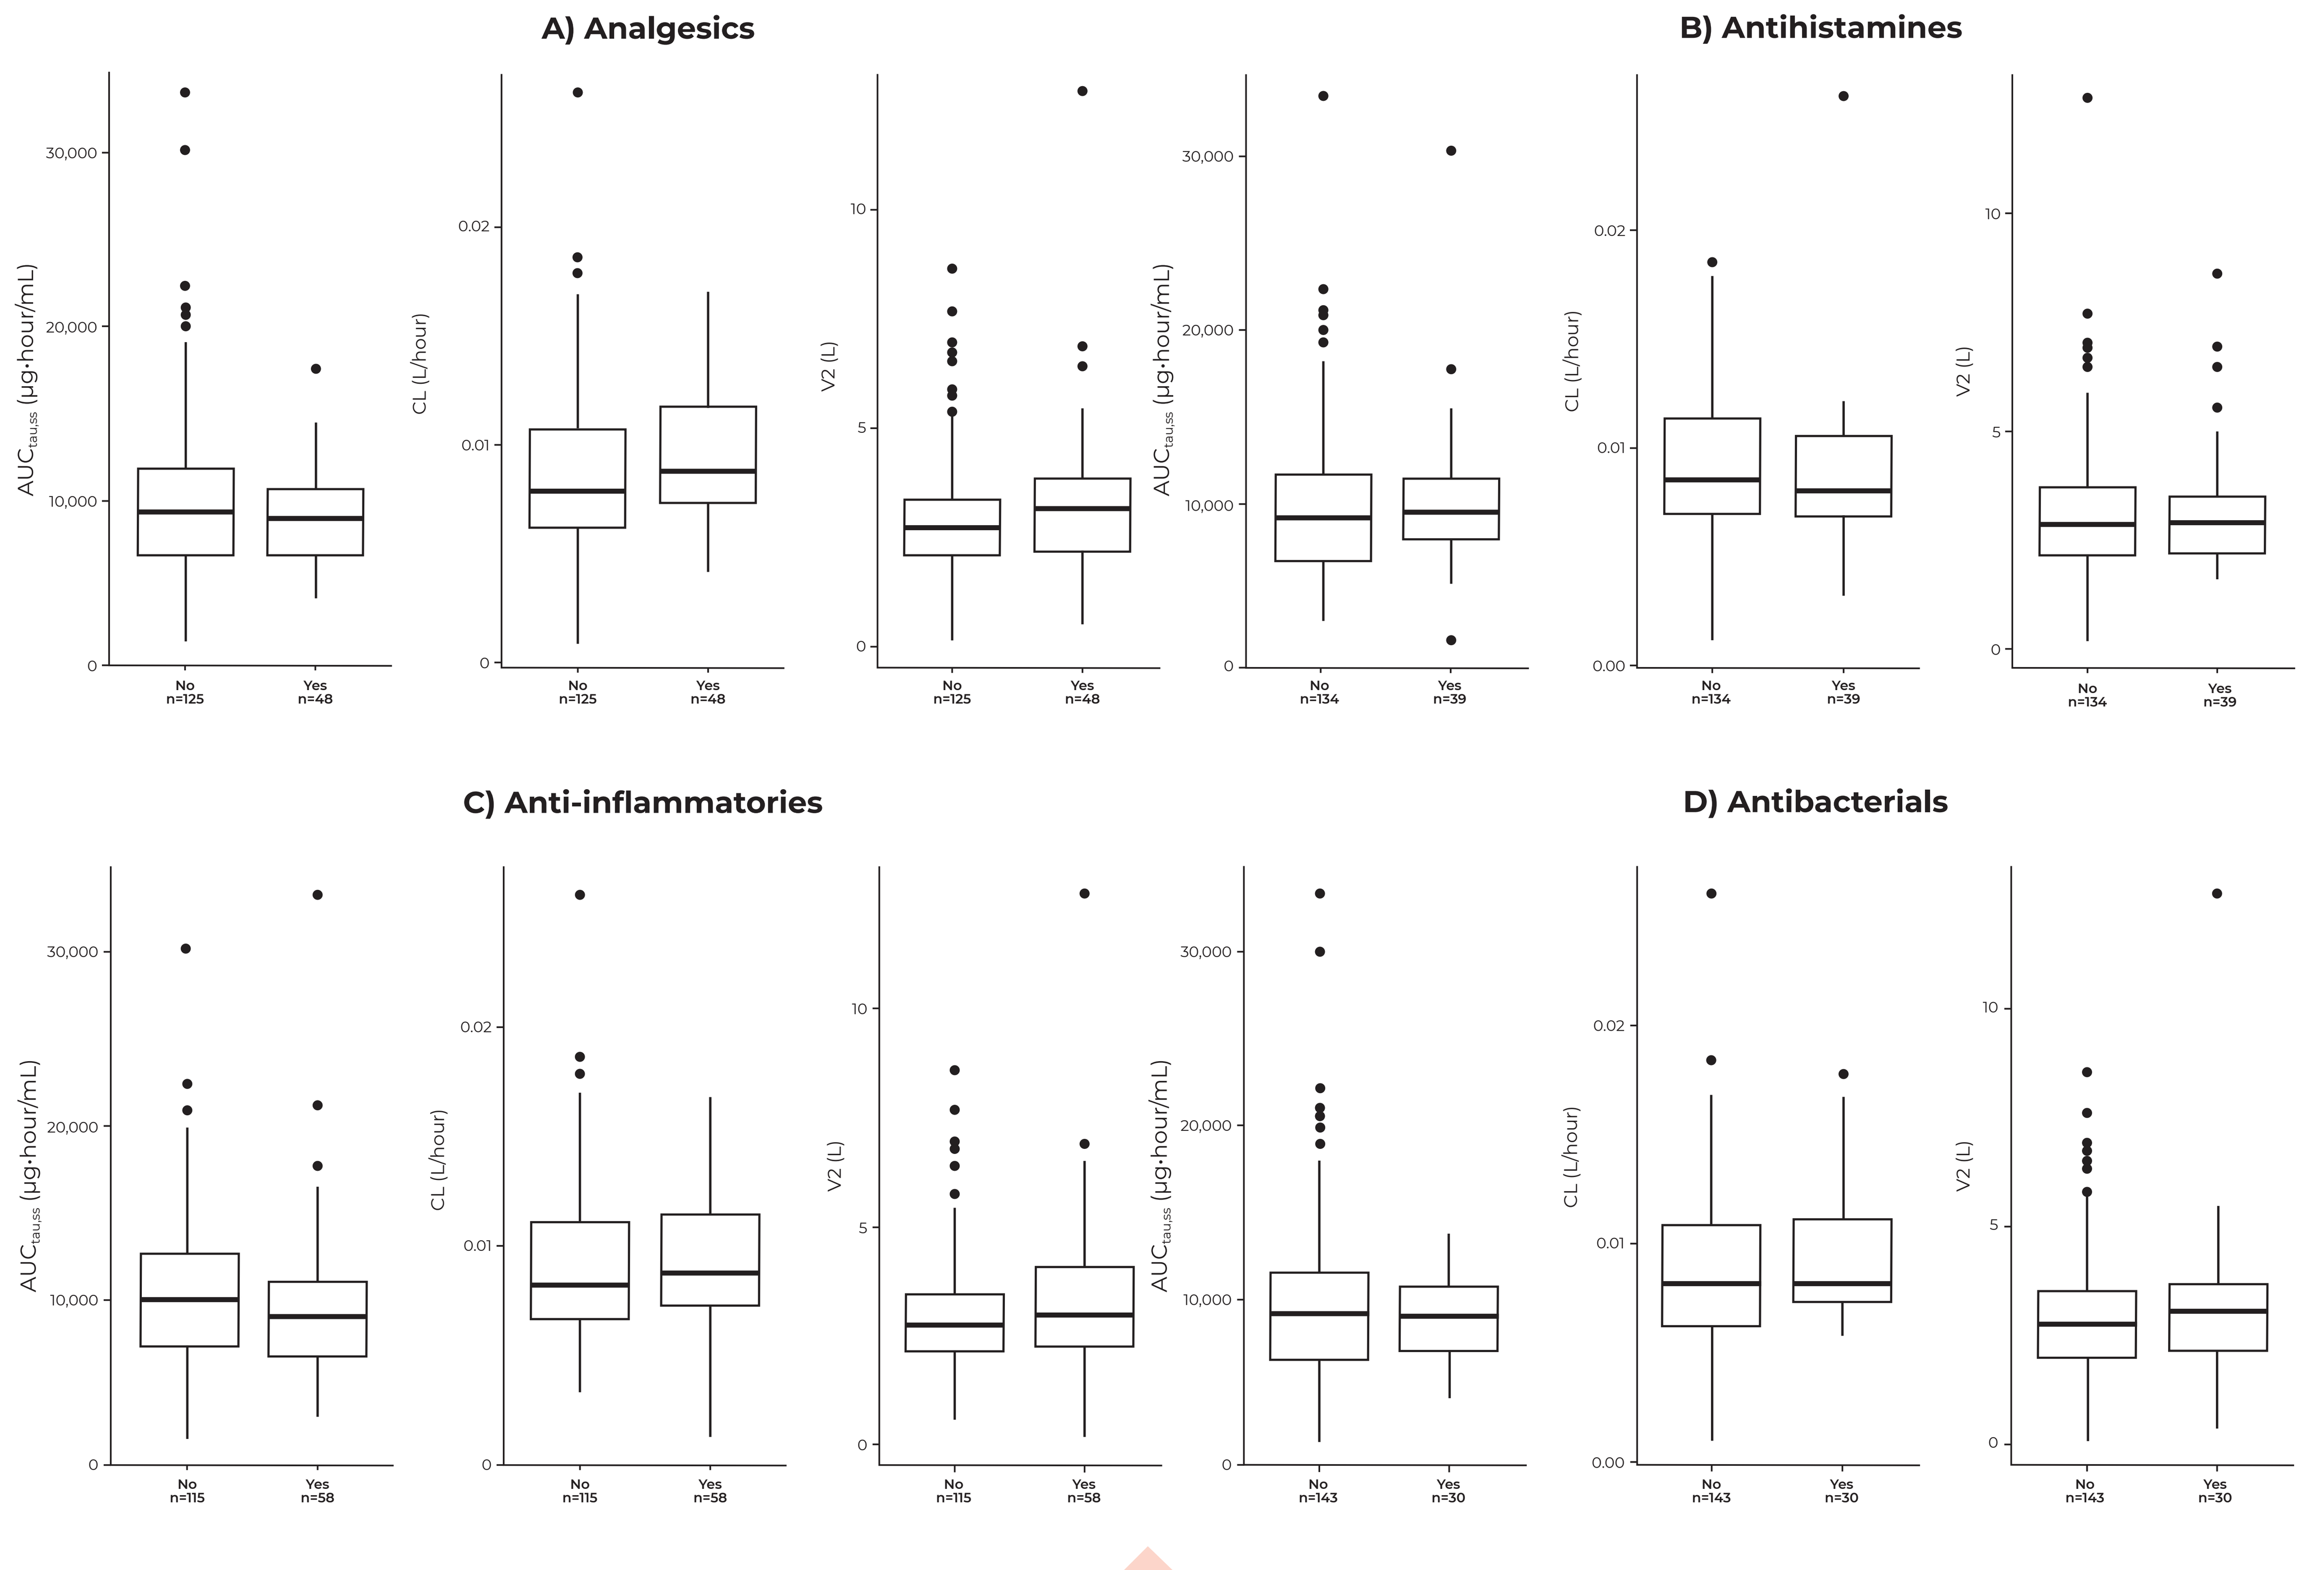
A) Analgesics
B) Antihistamines
C) Anti-inflammatories
D) Antibacterials
AUC$_{tau,ss}$ (µg·hour/mL)
CL (L/hour)
V2 (L)
No n=125
Yes n=48
No n=134
Yes n=39
No n=115
Yes n=58
No n=143
Yes n=30

Concomitant medications had **no impact** on the PK of garadacimab

In the box plots, median values are represented by a solid line in the center of the box. Boxes indicate the IQR, with whiskers extending to 1.5 × IQR.
AUC$_{tau,ss}$, area under the concentration–time curve for a dosing interval at steady state; CL, total drug clearance; HAE, hereditary angioedema; IQR, interquartile range; PK, pharmacokinetic; V2, volume of distribution.

DISCLOSURES:
This study was funded by CSL Behring LLC. Medical writing support was provided by Hazel Shepherd, DPhil, of Helix, OPEN Health Communications, and funded by CSL Behring, in accordance with Good Publication Practice guidelines (GPP 2022).

Scan the QR code to access the poster

American Academy of Allergy, Asthma, and Immunology/World Allergy Organization Joint Meeting | San Diego, CA, USA | February 28–March 3, 2025