Real-World Analysis of Treatment Patterns Among Patients With Hereditary Angioedema in the United States

L12

Raffi Tachdjian[1], Chiara Nenci[2], Vai Katkade[3], Darcy Ellis[3], Yinglei Li[3], Abisola Olopoenia[4], Marc A. Riedl[5]

[1]Division of Allergy and Clinical Immunology, David Geffen School of Medicine, University of California, Los Angeles, Los Angeles, CA, USA; [2]CSL Behring AG, Bern, Switzerland; [3]CSL Behring, King of Prussia, PA, USA; [4]Oracle Corporation, Redwood City, CA, USA; [5]University of California San Diego, San Diego, CA, USA.

CONCLUSIONS

- **A substantial proportion of patients received on-demand treatment (ODT) as index therapy, many of whom initiated long-term prophylaxis (LTP) within 12 months of follow-up**
- **Most patients who received index LTP continued LTP, with 6.8% switching to another LTP**
- **The majority of LTP-to-LTP switches were made within 5 weeks, and one-third of these patients switched multiple times**
- **These data suggest that hereditary angioedema (HAE) treatment is being continually optimized in the USA, particularly with the use of LTP**

BACKGROUND

- Multiple therapeutic options are available and recommended for the treatment or prevention of HAE attacks, including ODT and LTP[1,2]
- The US HAEA Medical Advisory Board guidelines were updated in 2020,[1] and WAO/EAACI guidelines in 2021, both of which noted that key treatment goals can only be achieved with effective LTP[2]
- The most recent evaluation of real-world treatment patterns in the USA was published in 2018 (using data collected between 2006 and 2014), before the introduction of several targeted LTP options, and before treatment guidelines were updated[3–6]
- As new treatment options are introduced, comprehensive understanding of treatment utilization and switching is needed to inform future use of new treatments by identifying and addressing unmet needs in a real-world setting

OBJECTIVE

- To provide a comprehensive evaluation of current HAE-specific treatment patterns using US administrative claims from 2018 to 2024

METHODS AND PATIENT DISTRIBUTION

- The Oracle Life Sciences database included prescription and medical claims data for 2867 patients with HAE, with a range of 30 days–6.5 years of continuous insurance coverage from July 1, 2018 to March 31, 2024
- Index therapy (medication received at the start of follow-up) and baseline characteristics from medical and pharmacy claims data were recorded in the database
- Treatment patterns (continuation, discontinuation, suspension, LTP initiation, and LTP switch) were analyzed and expressed using descriptive statistics
- The proportion of patients who received index therapy or changed therapies was assessed over 12 months for patients with sufficient, continuous post-index insurance coverage

Patient distribution

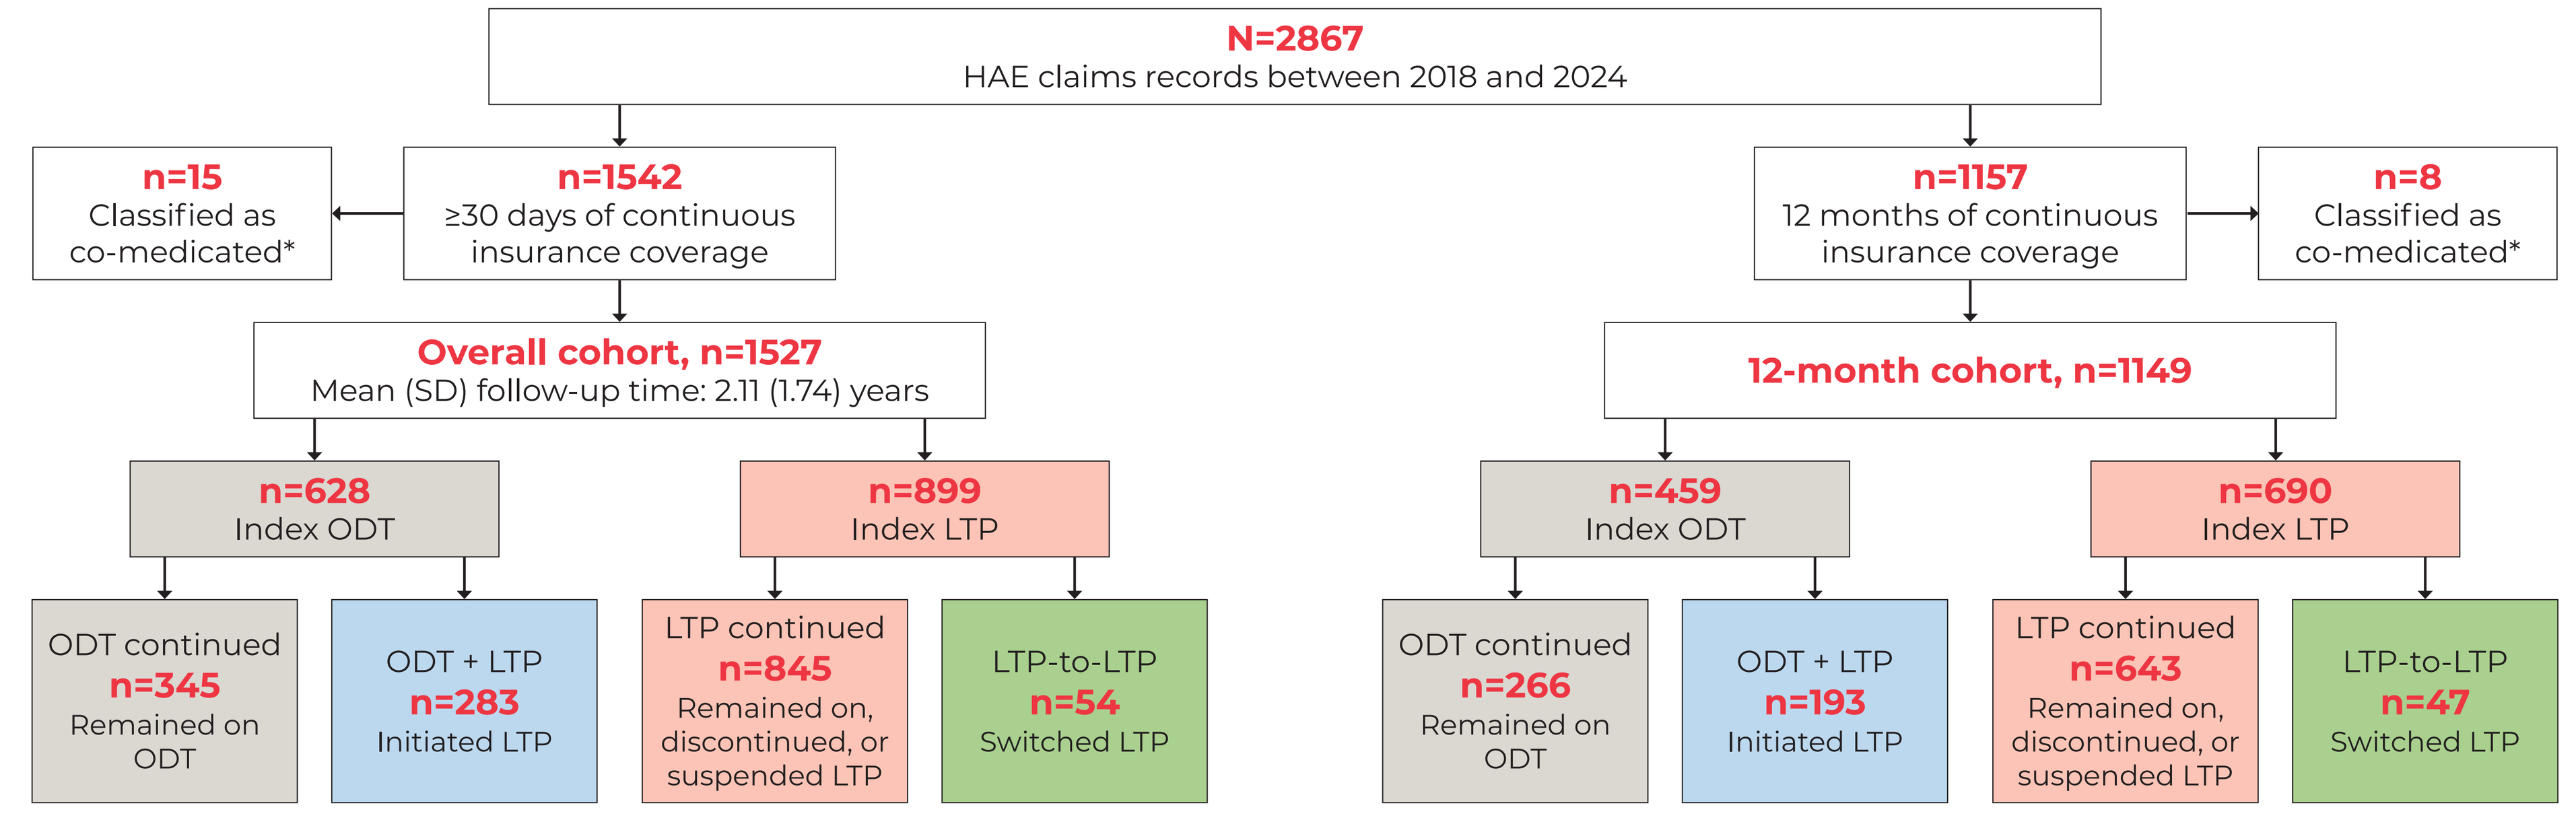
N=2867
HAE claims records between 2018 and 2024
n=15
Classified as co-medicated*
n=1542
≥30 days of continuous insurance coverage
n=1157
12 months of continuous insurance coverage
n=8
Classified as co-medicated*
Overall cohort, n=1527
Mean (SD) follow-up time: 2.11 (1.74) years
12-month cohort, n=1149
n=628
Index ODT
n=899
Index LTP
n=459
Index ODT
n=690
Index LTP
ODT continued
n=345
Remained on ODT
ODT + LTP
n=283
Initiated LTP
LTP continued
n=845
Remained on, discontinued, or suspended LTP
LTP-to-LTP
n=54
Switched LTP
ODT continued
n=266
Remained on ODT
ODT + LTP
n=193
Initiated LTP
LTP continued
n=643
Remained on, discontinued, or suspended LTP
LTP-to-LTP
n=47
Switched LTP

Baseline demographics and characteristics

Characteristic	Overall cohort (n=1527)	12-month cohort (n=1149)
Mean age as of index date, years (SD)	37.1 (17.2)	37.5 (17.2)
Sex, female, %	70.9	71.9
Payer type, %		
Commercial	48.1	49.4
Medicaid managed care	38.7	37.6
Medicare Advantage	5.5	5.4
Self-insured	5.7	6.1
Other/unknown	2.0	1.5

*Patients received two LTP therapies concurrently and were not included in this analysis.
EAACI, European Academy of Allergy and Clinical Immunology; HAE, hereditary angioedema; HAEA, Hereditary Angioedema Association; LTP, long-term prophylaxis; ODT, on-demand treatment; SD, standard deviation; WAO, World Allergy Organization.

RESULTS

A large proportion of patients originally received ODT

Figure 1. Index therapy for patients with ≥30 days of continuous insurance coverage

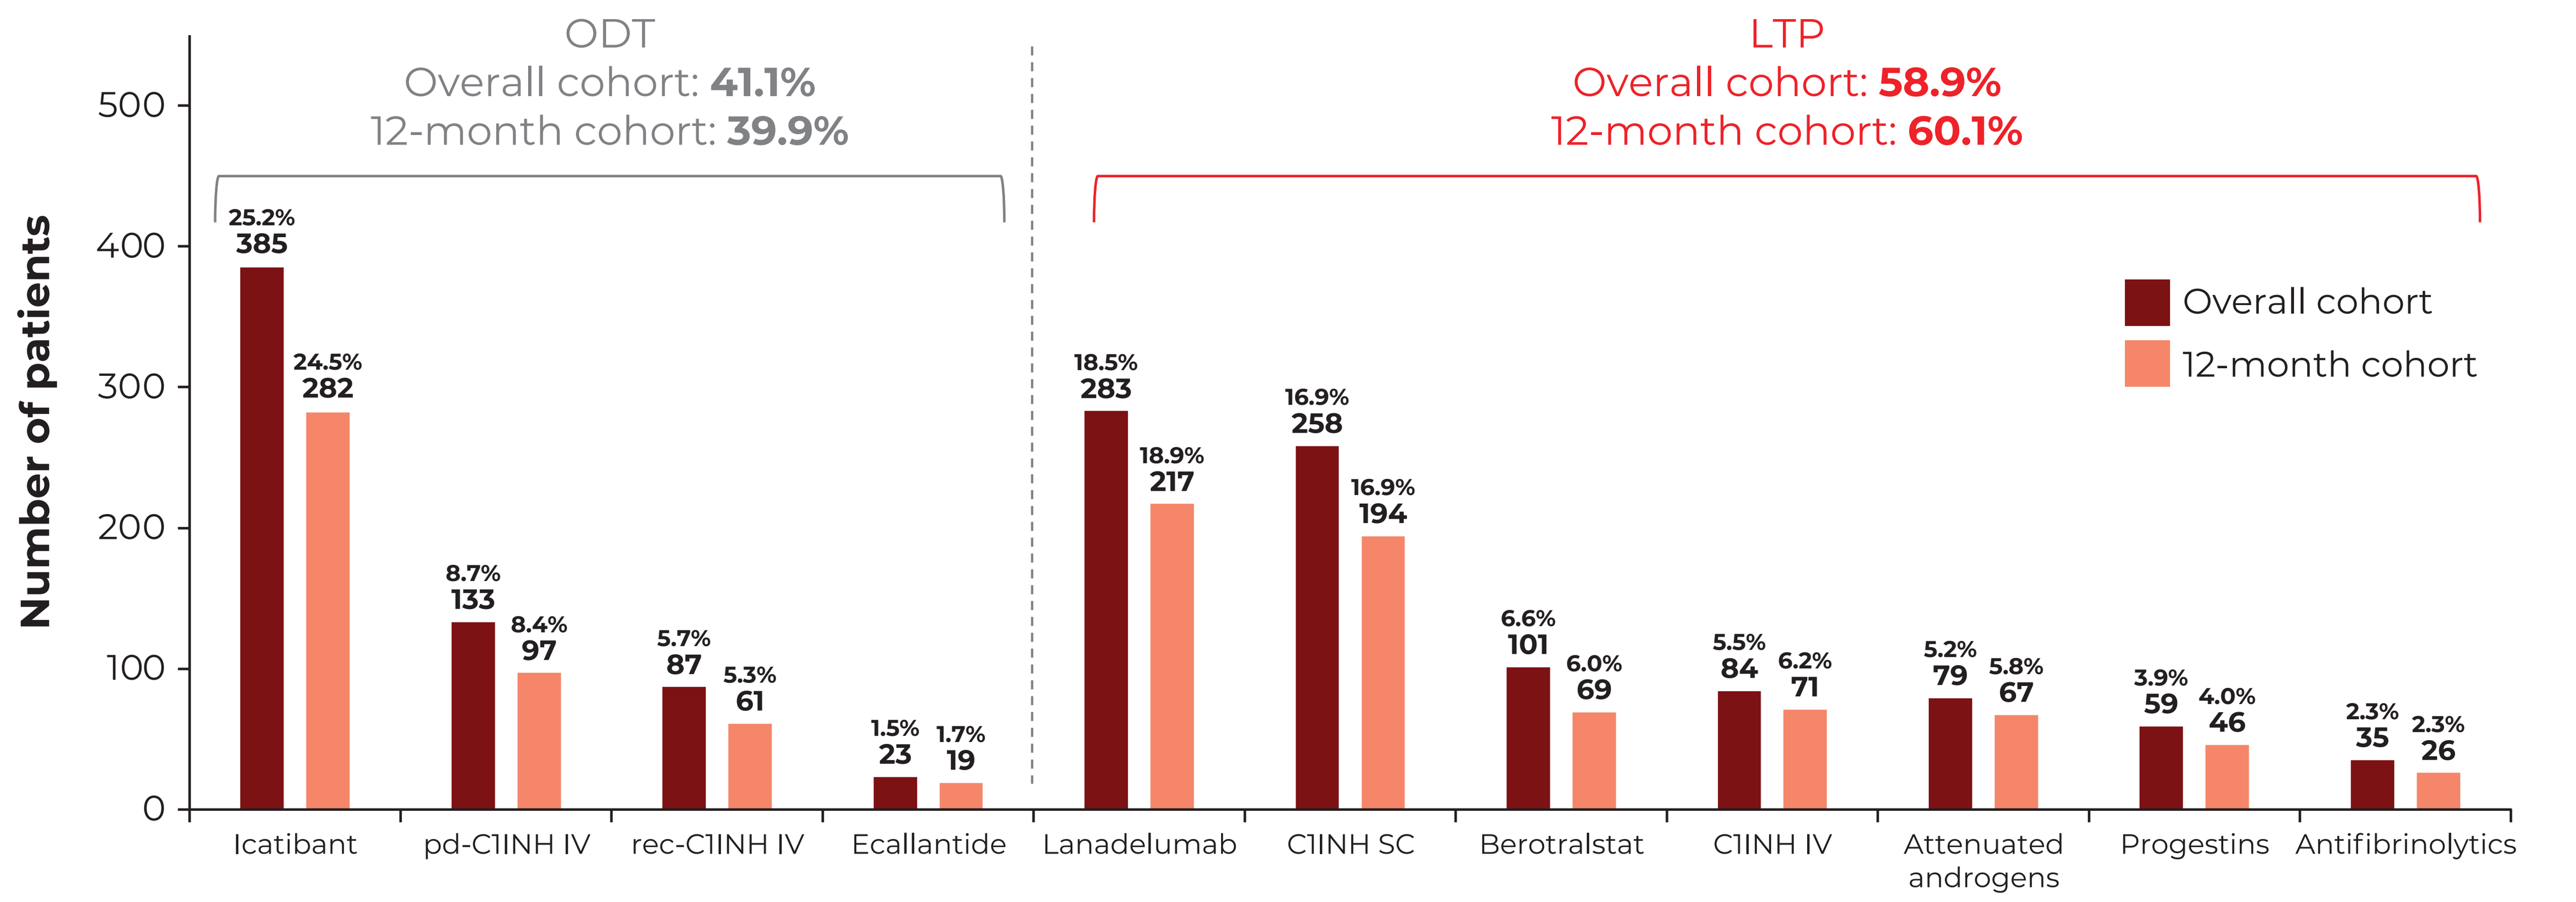
ODT
Overall cohort: 41.1%
12-month cohort: 39.9%
LTP
Overall cohort: 58.9%
12-month cohort: 60.1%
Number of patients
Overall cohort
12-month cohort
25.2% 385
24.5% 282
8.7% 133
8.4% 97
5.7% 87
5.3% 61
1.5% 23
1.7% 19
18.5% 283
18.9% 217
16.9% 258
16.9% 194
6.6% 101
6.0% 69
5.5% 84
6.2% 71
5.2% 79
5.8% 67
3.9% 59
4.0% 46
2.3% 35
2.3% 26
Icatibant
pd-C1INH IV
rec-C1INH IV
Ecallantide
Lanadelumab
C1INH SC
Berotralstat
C1INH IV
Attenuated androgens
Progestins
Antifibrinolytics

Proportion of patients in each cohort who received index HAE therapy in each study year

Patient cohort	2018	2019	2020	2021	2022	2023	2024
Overall cohort, %	36.0	15.0	7.7	12.0	14.6	12.2	2.5
12-month cohort, %	37.2	14.8	8.7	12.9	16.3	9.2	1.0

C1INH, C1 inhibitor; HAE, hereditary angioedema; IV, intravenous; LTP, long-term prophylaxis; ODT, on-demand treatment; pd, plasma-derived; rec, recombinant human; SC, subcutaneous.

Patients who received LTP were more likely to remain on index therapy than those who received ODT

Figure 2. Treatment patterns in the 12-month cohort (n=1149)

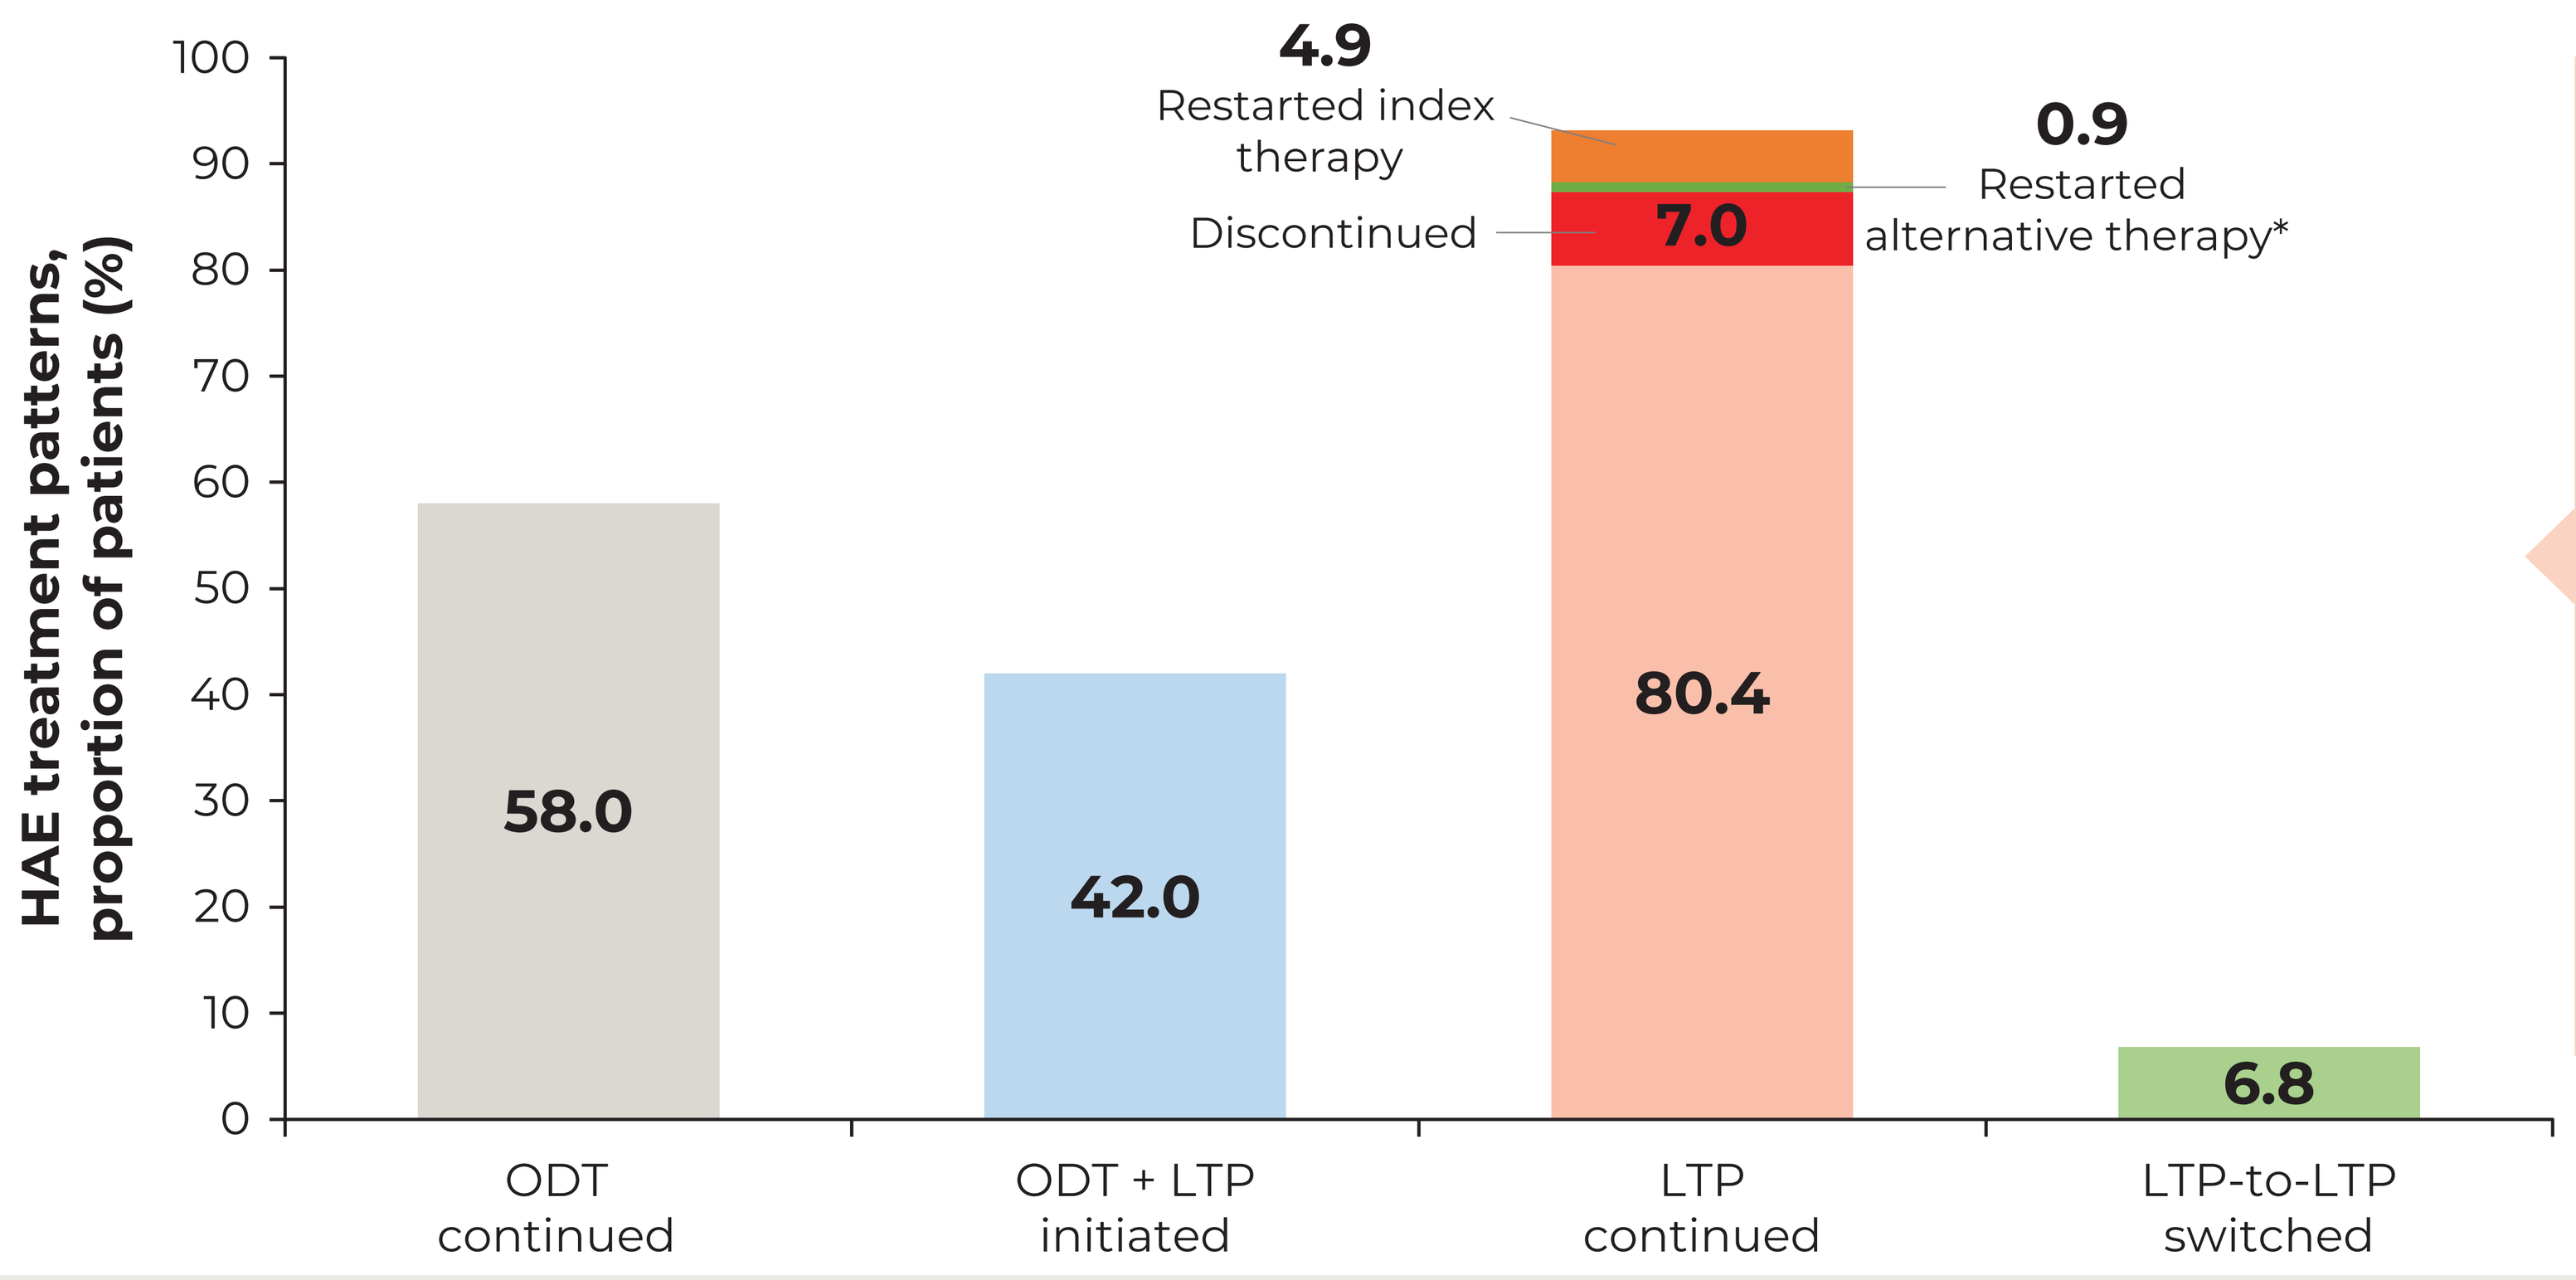
HAE treatment patterns, proportion of patients (%)
58.0
42.0
80.4
7.0
4.9
Restarted index therapy
Discontinued
0.9
Restarted alternative therapy*
6.8
ODT continued
ODT + LTP initiated
LTP continued
LTP-to-LTP switched

Among LTP continuers, 88 (12.8%) discontinued treatment, of whom **34 (38.6%) restarted their index LTP** and six (6.8%) restarted an alternative LTP

*Of those who restarted an alternative LTP, index LTP therapies were attenuated androgens (n=2), C1INH SC (n=2), progestins (n=1), and lanadelumab (n=1).
C1INH, C1 inhibitor; LTP, long-term prophylaxis; ODT, on-demand treatment; SC, subcutaneous.

Figure 3. The majority of patients who initiated LTP from ODT initiated C1INH SC or lanadelumab; most patients who switched LTP-to-LTP switched from antifibrinolytics or attenuated androgens

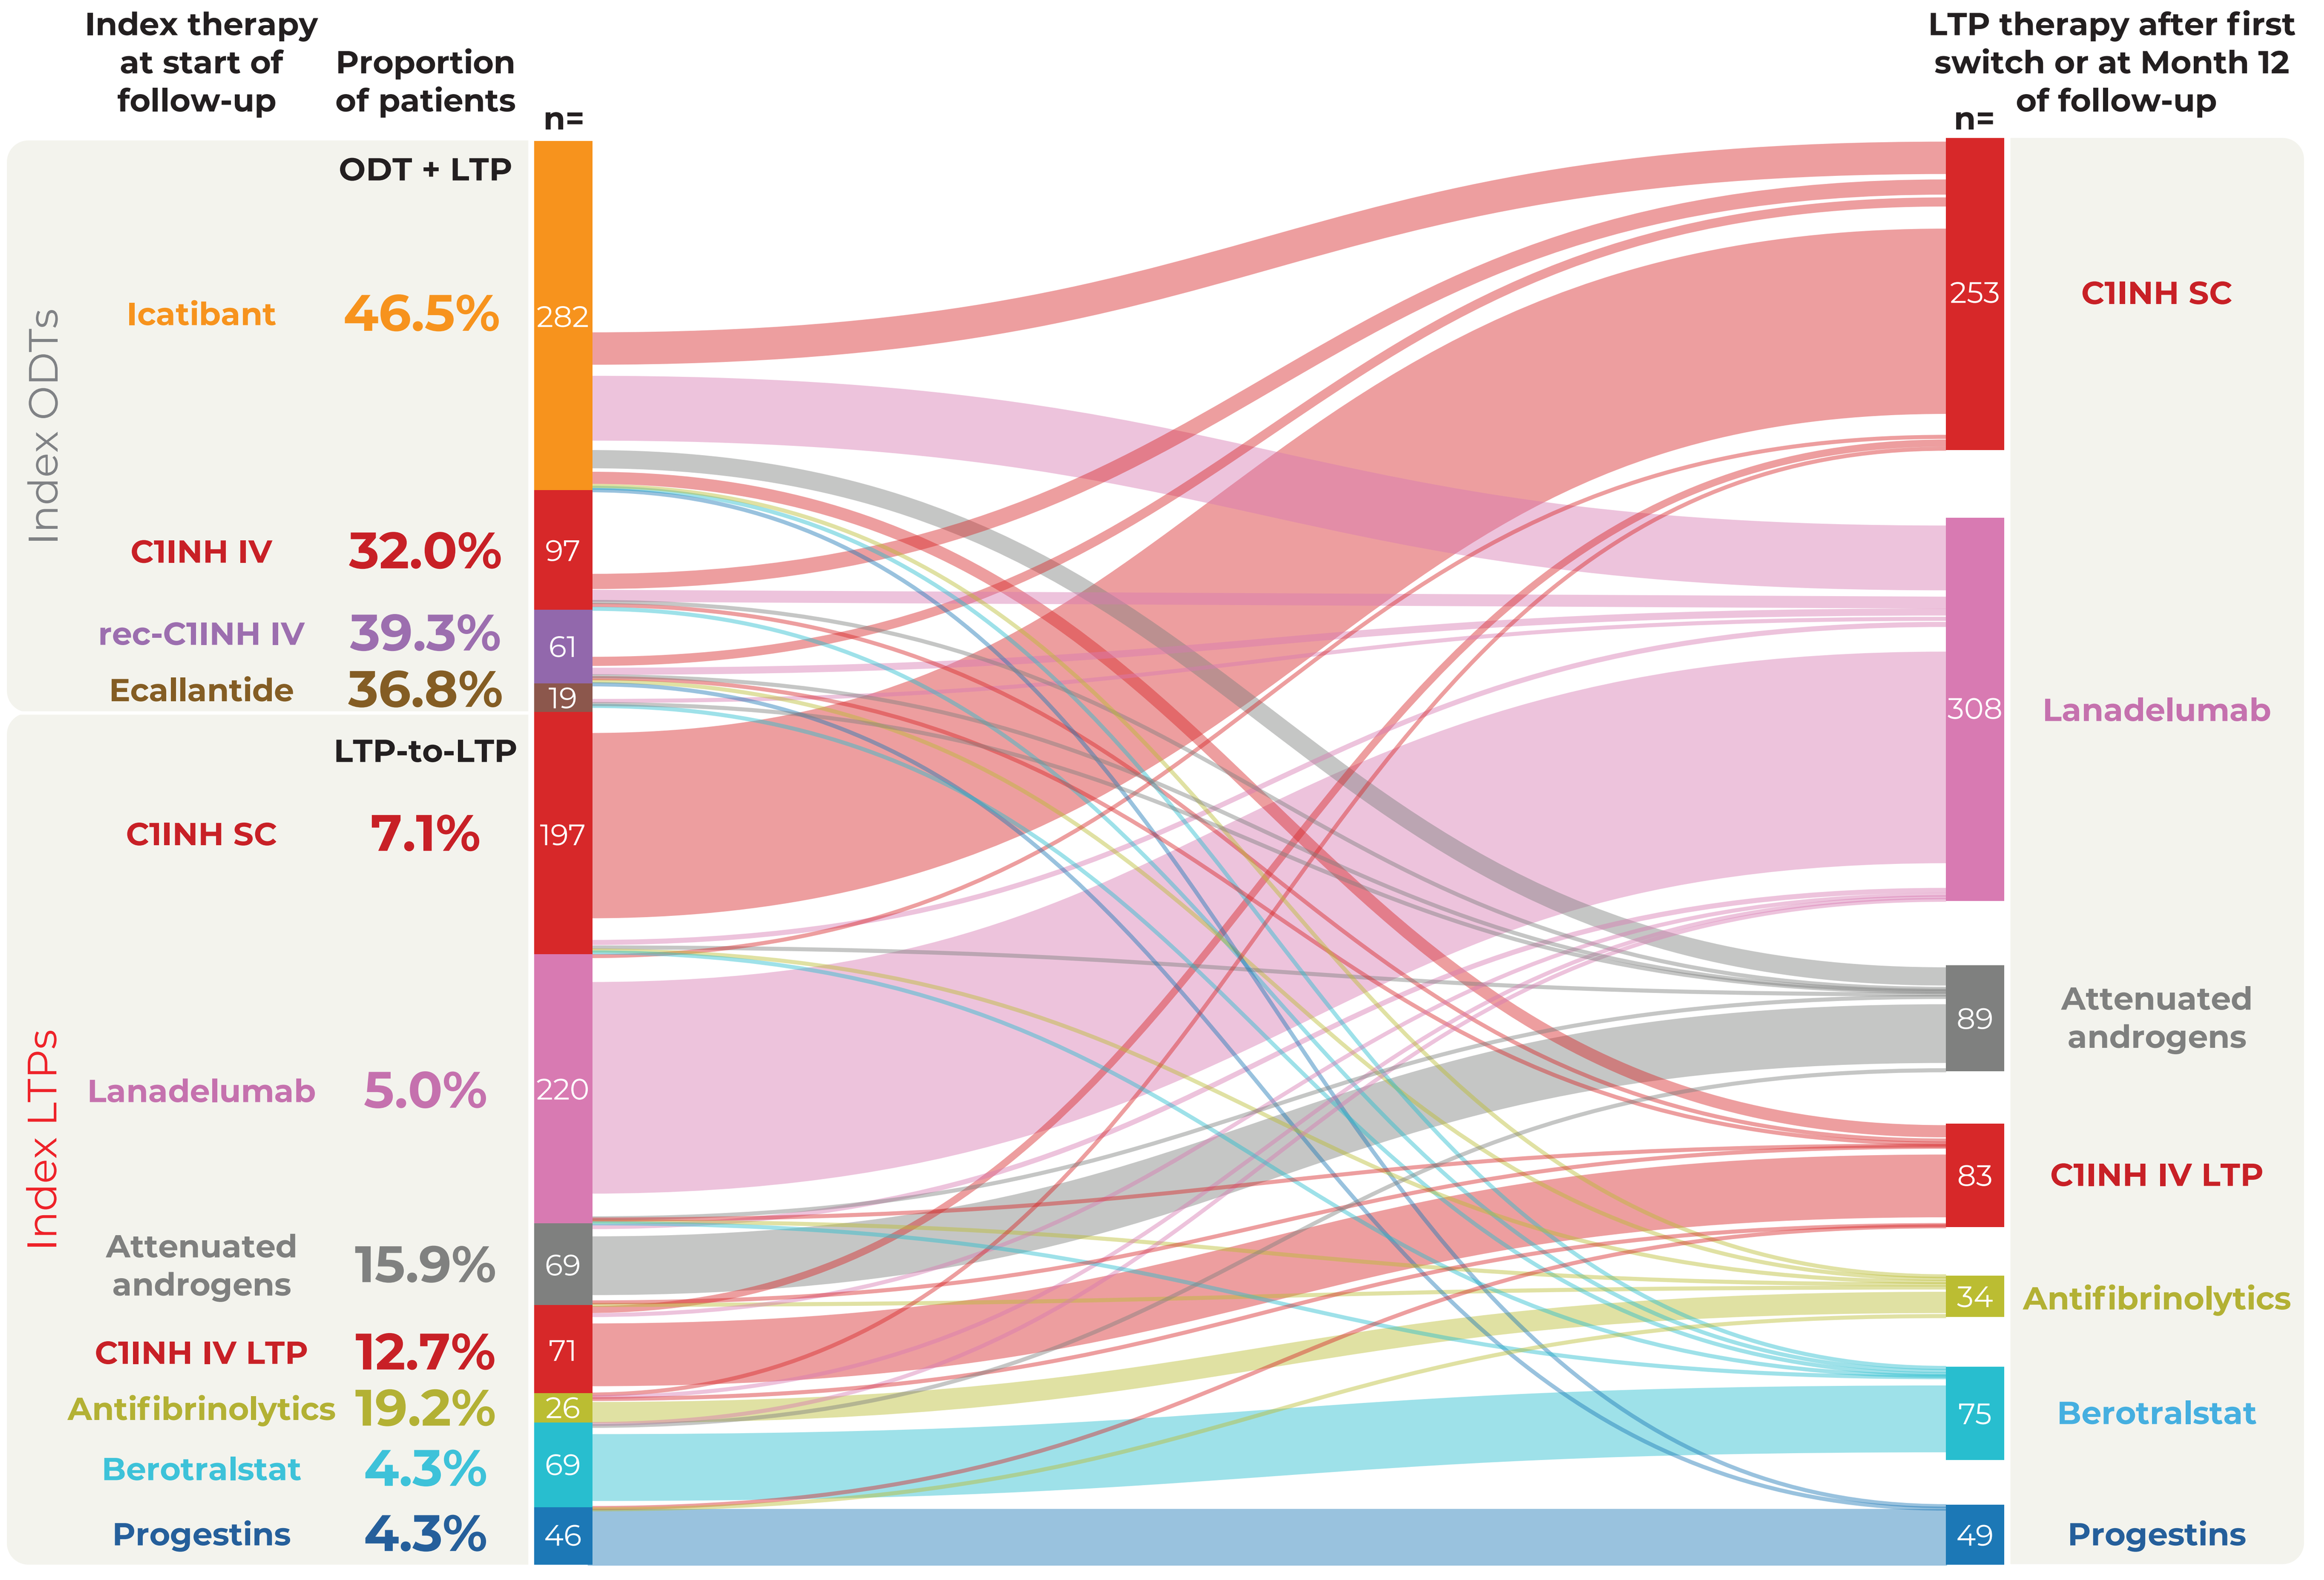
Index therapy at start of follow-up
Proportion of patients
n=
LTP therapy after first switch or at Month 12 of follow-up
n=
Index ODTs
ODT + LTP
Icatibant 46.5% 282
C1INH IV 32.0% 97
rec-C1INH IV 39.3% 61
Ecallantide 36.8% 19
Index LTPs
LTP-to-LTP
C1INH SC 7.1% 197
Lanadelumab 5.0% 220
Attenuated androgens 15.9% 69
C1INH IV LTP 12.7% 71
Antifibrinolytics 19.2% 26
Berotralstat 4.3% 69
Progestins 4.3% 46
253 C1INH SC
308 Lanadelumab
89 Attenuated androgens
83 C1INH IV LTP
34 Antifibrinolytics
75 Berotralstat
49 Progestins

- Mean (SD) time to first LTP-to-LTP switch was 1.88 (1.66) months
- Multiple LTP-to-LTP switches were recorded for 14 (29.8%) of the 47 patients who switched (mean [SD]: 2.21 [0.43] per patient)

Of patients who received index ODT, **42.0% initiated LTP** within the 12-month follow-up.
Of patients who received index LTP, **6.8% switched to another LTP**

C1INH, C1 inhibitor; IV, intravenous; LTP, long-term prophylaxis; ODT, on-demand treatment; SC, subcutaneous; SD, standard deviation.

Scan the QR code to access the poster

REFERENCES:
1. Busse PJ et al. *J Allergy Clin Immunol Pract* 2021;9:132–150.e3; 2. Maurer M et al. *Allergy* 2022;77:1961–1990;
3. Riedl MA et al. *Orphanet J Rare Dis* 2018;13:180; 4. Takeda. TAKHZYRO (lanadelumab) Prescribing Information. 2023;
5. CSL Behring. HAEGARDA (C1INH SC) Prescribing Information. 2022; 6. BioCryst. ORLADEYO (berotralstat) Prescribing Information. 2020.

DISCLOSURES:
This study was funded by CSL Behring LLC. Medical writing support was provided by Hazel Shepherd, DPhil, of Helix, OPEN Health Communications, and funded by CSL Behring, in accordance with Good Publication Practice guidelines (GPP 2022).

American Academy of Allergy, Asthma, and Immunology/World Allergy Organization Joint Meeting | San Diego, CA, USA | February 28–March 3, 2025