P264

Exploring patient preferences for self-administered subcutaneous therapy in chronic inflammatory demyelinating polyneuropathy

Batyrkhan Kuatov,[1] Sarah-Jane Cashmore,[2] Noemi Hahn,[2] Julia Braverman,[3] Laurence Undreiner,[4] Shicheng Weng,[3] Ben Gibbons,[2] Jane Esman[5]

1. CSL, Bern, Switzerland
2. Bryter Inc., New York, NY, United States
3. CSL, Waltham, MA, USA
4. CSL, Paris, France
5. CSL, Plymouth, MA, USA

Conclusions

- Patient choice for self-administered SCIg in CIDP appears to be driven more by preparation simplicity and manageable injection volumes than by extending dosing intervals alone
- SCIg PFS presentation aligns closely with key patient preference drivers for self-administered SCIg therapy compared with vial presentation
- Ready-to-use, low-burden delivery formats such as PFS may enhance treatment acceptability and support self-administration, although further research is needed to confirm effects on adherence

Introduction

- Chronic inflammatory demyelinating polyneuropathy (CIDP) is an autoimmune disease affecting the peripheral nerve system.[1] CIDP results in sensory and reflex loss, fatigue, and a slow symmetrical onset of relapsing or progressive proximal limb weakness[2,3]
- Immunoglobulin (Ig) therapy is a cornerstone of CIDP management.[4] Patients often transition from intravenous immunoglobulin (IVIg) induction to subcutaneous immunoglobulin (SCIg) for maintenance, which can be administered at home with a reduction in infusion times[4,5]
- SCIg is available in vial-based formats and ready-to-use prefilled syringes (PFS), with differences in preparation complexity and physical handling requirements.[6] However, direct comparative evidence on SCIg packaging formats in CIDP is limited
- As SCIg use expands, practical aspects of home administration, such as preparation steps, handling burden, and infusion setup, are increasingly important determinants of patient preference. Patient preferences should be considered in CIDP treatment selection to support shared decision-making[4]

Objective

This study aimed to identify the key treatment attributes that influence patient preferences for self-administered subcutaneous therapy in CIDP

Methods

- Eligible patients with CIDP using Ig from the United States and Canada completed an online 30-minute questionnaire, which included a conjoint exercise
- Eligibility was determined through a structured screening questionnaire that assessed the following inclusion criteria: 18 years of age or older; have undergone at least one diagnostic procedure for CIDP; possess a physician-confirmed CIDP diagnosis; have their diagnosis established or current care managed by either a neurologist or neuromuscular disease specialist; be actively receiving either IVIg or SCIg therapy for CIDP management; and have maintained continuous Ig treatment for a minimum duration of 6 months
- The conjoint exercise was conducted with Ig-using patients with CIDP to determine the relative importance of various treatment attributes in self-administered subcutaneous therapy
- In addition, survey questions with SCIg-using patients with CIDP evaluated satisfaction and preferences between prefilled syringes (PFS) and vials

Figure 1. Patient perceptions of the impact of switching SCIg packaging from vials to PFS (n = 17) on (A) Treatment satisfaction, (B) Convenience of administering infusion and (C) Infusion time

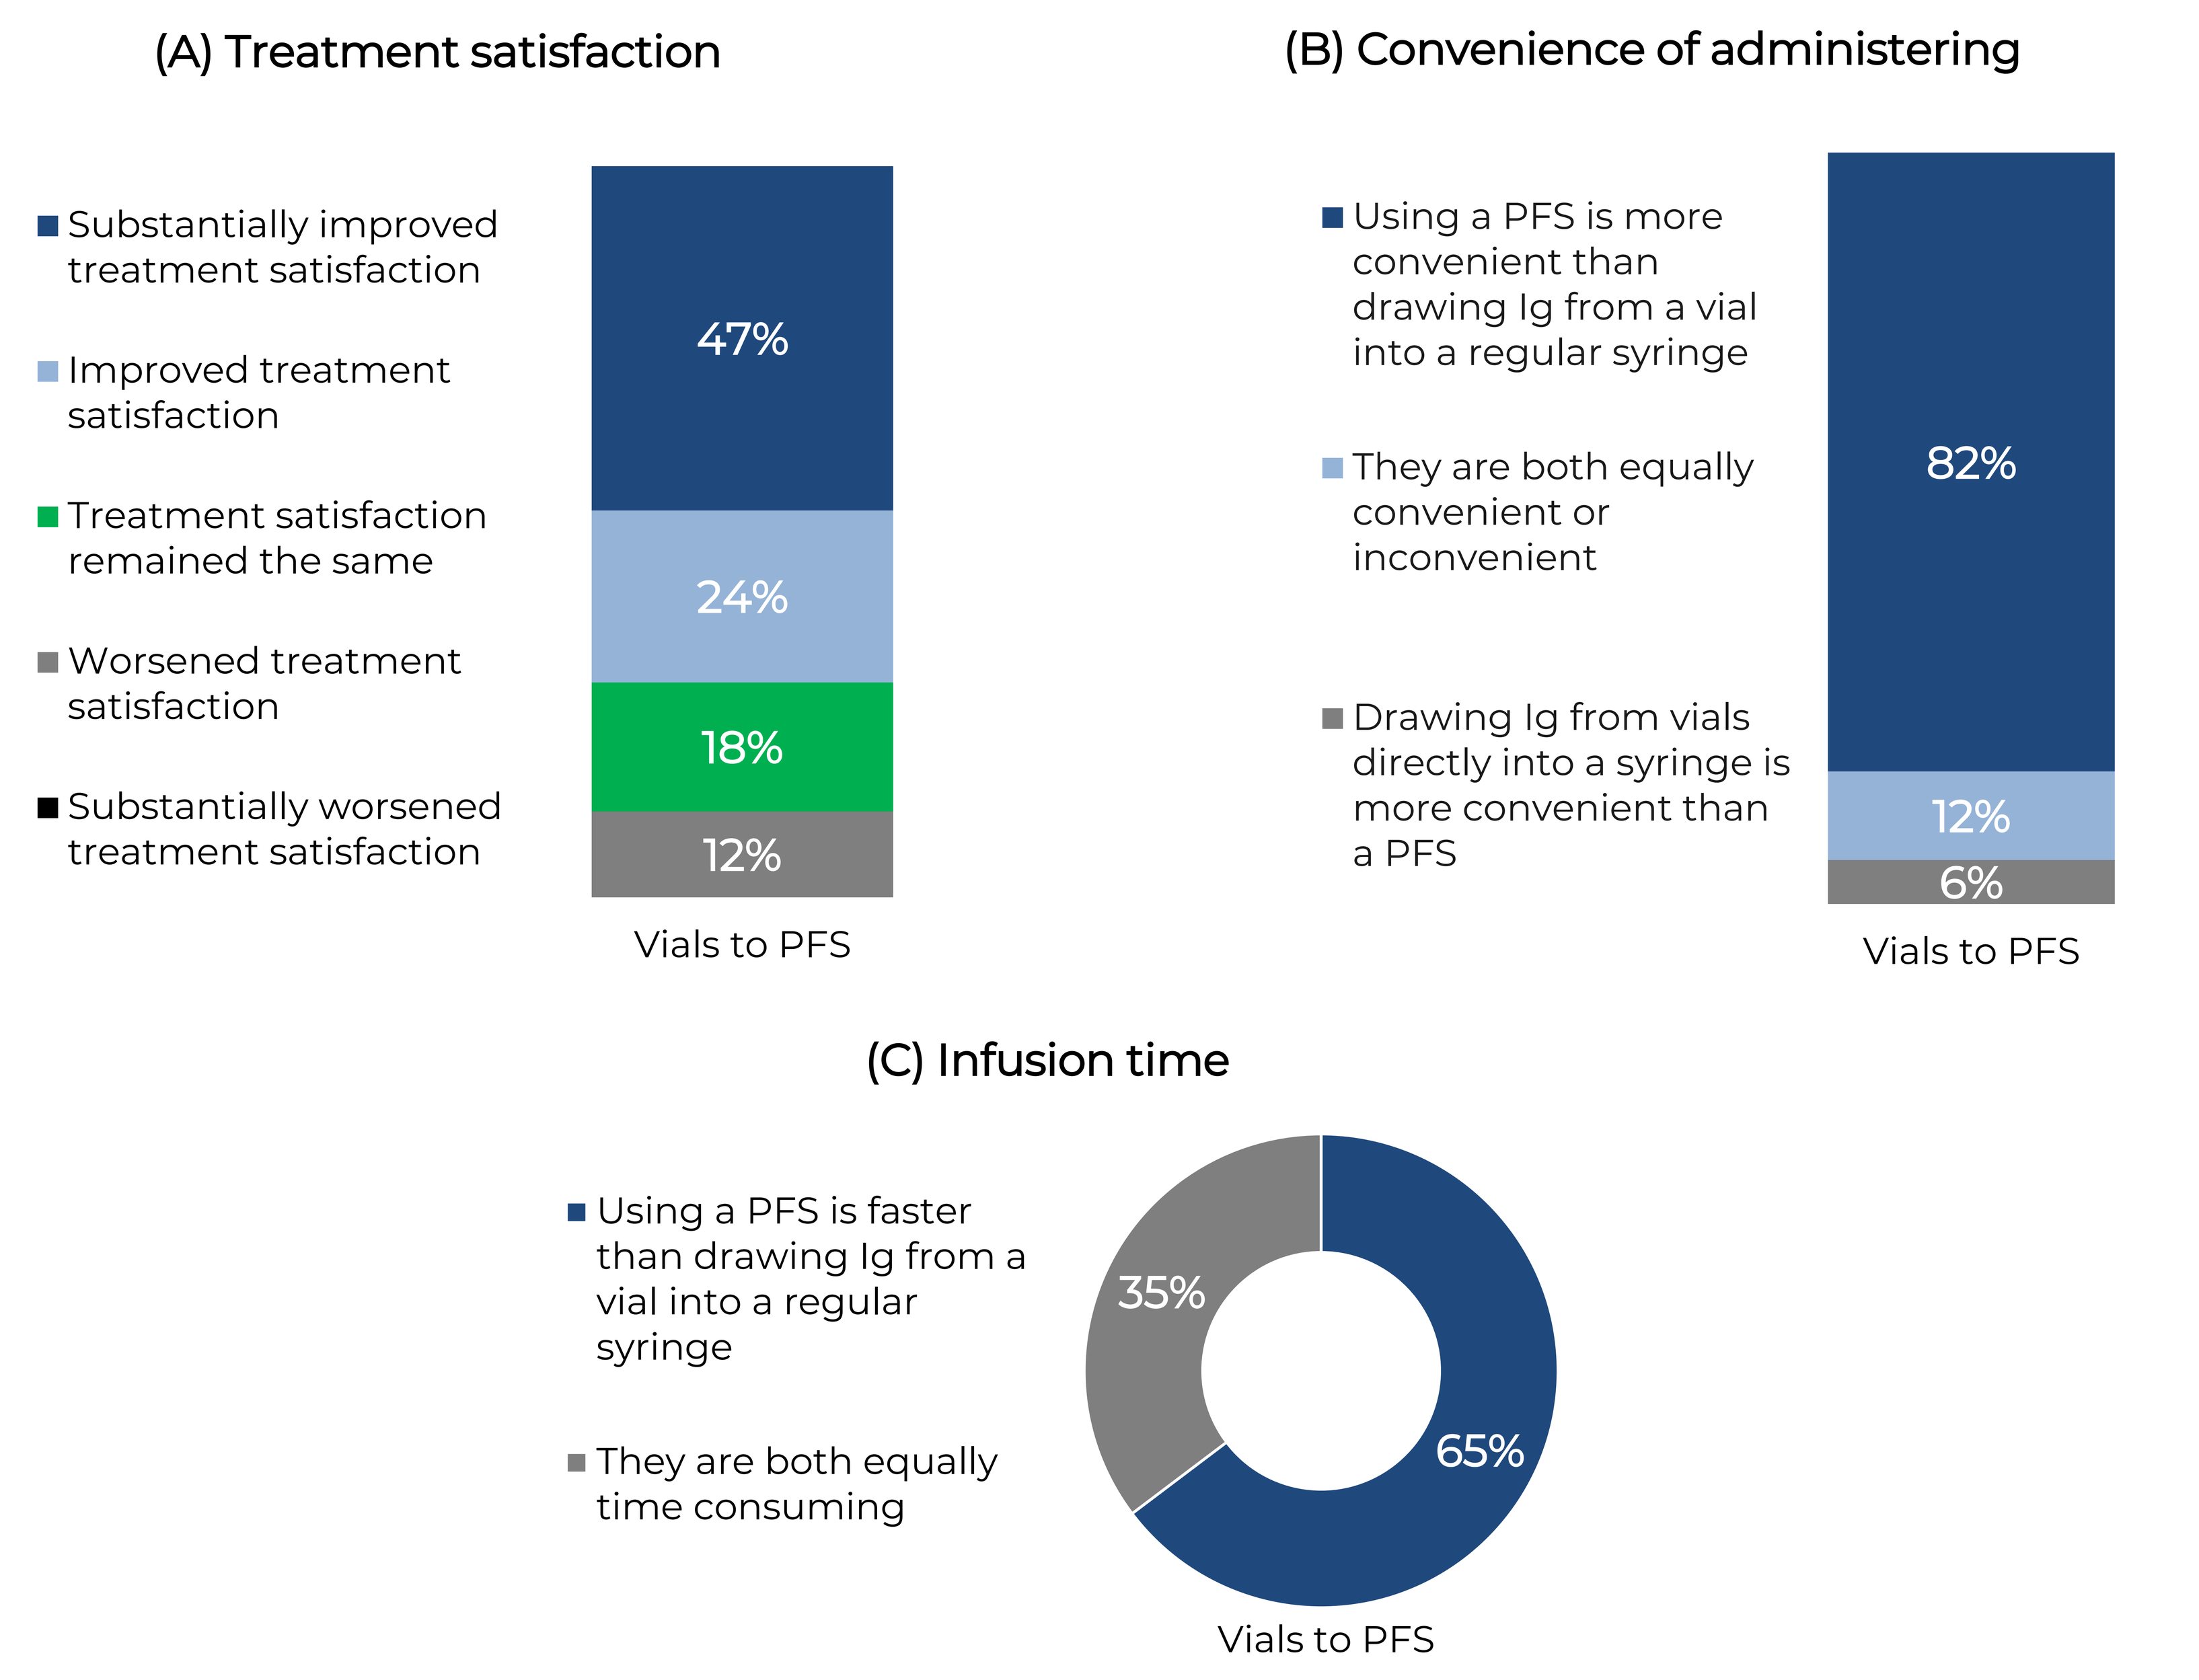
(A) Treatment satisfaction
Substantially improved treatment satisfaction
Improved treatment satisfaction
Treatment satisfaction remained the same
Worsened treatment satisfaction
Substantially worsened treatment satisfaction
47%
24%
18%
12%
Vials to PFS
(B) Convenience of administering
Using a PFS is more convenient than drawing Ig from a vial into a regular syringe
They are both equally convenient or inconvenient
Drawing Ig from vials directly into a syringe is more convenient than a PFS
82%
12%
6%
Vials to PFS
(C) Infusion time
Using a PFS is faster than drawing Ig from a vial into a regular syringe
They are both equally time consuming
35%
65%
Vials to PFS

Ig, immunoglobulin; PFS, prefilled syringe; SCIg, subcutaneous immunoglobulin

Results

- A total of 187 patients with CIDP were enrolled in this study, comprising 150 patients from the United States and 37 from Canada. The mean age of the cohort was 55 years and there were more female patients (65%) than male (35%). At enrollment, 119 patients were receiving IVIg and 68 were receiving SCIg (PFS users n = 43, vial users n = 25)
- Of the SCIg patients who switched from vials to PFS (n = 17), 71% reported improved satisfaction with their treatment, 82% thought PFS was more convenient than vials, and 65% believed that PFS administration was faster than using a vial (**Figure 1**)
- The conjoint analysis determined that ease of preparation (31% of relative importance) and likelihood of getting sick after infusion (27% of relative importance) were the main drivers of preference in subcutaneous therapies. Injection volume and skin reaction (e.g., swelling) ranked third (23% of relative importance) (**Figure 2**)
- When surveyed, PFS users reported feeling more confident (completely/very confident) administering their infusions than vial-users (**Figure 3**)
- PFS users felt better than vial users within the first day after an infusion (51% vs 20% felt 'better than usual', respectively) (**Figure 4**)

Figure 2. Overall Relative Attribute Importance score (RAI)

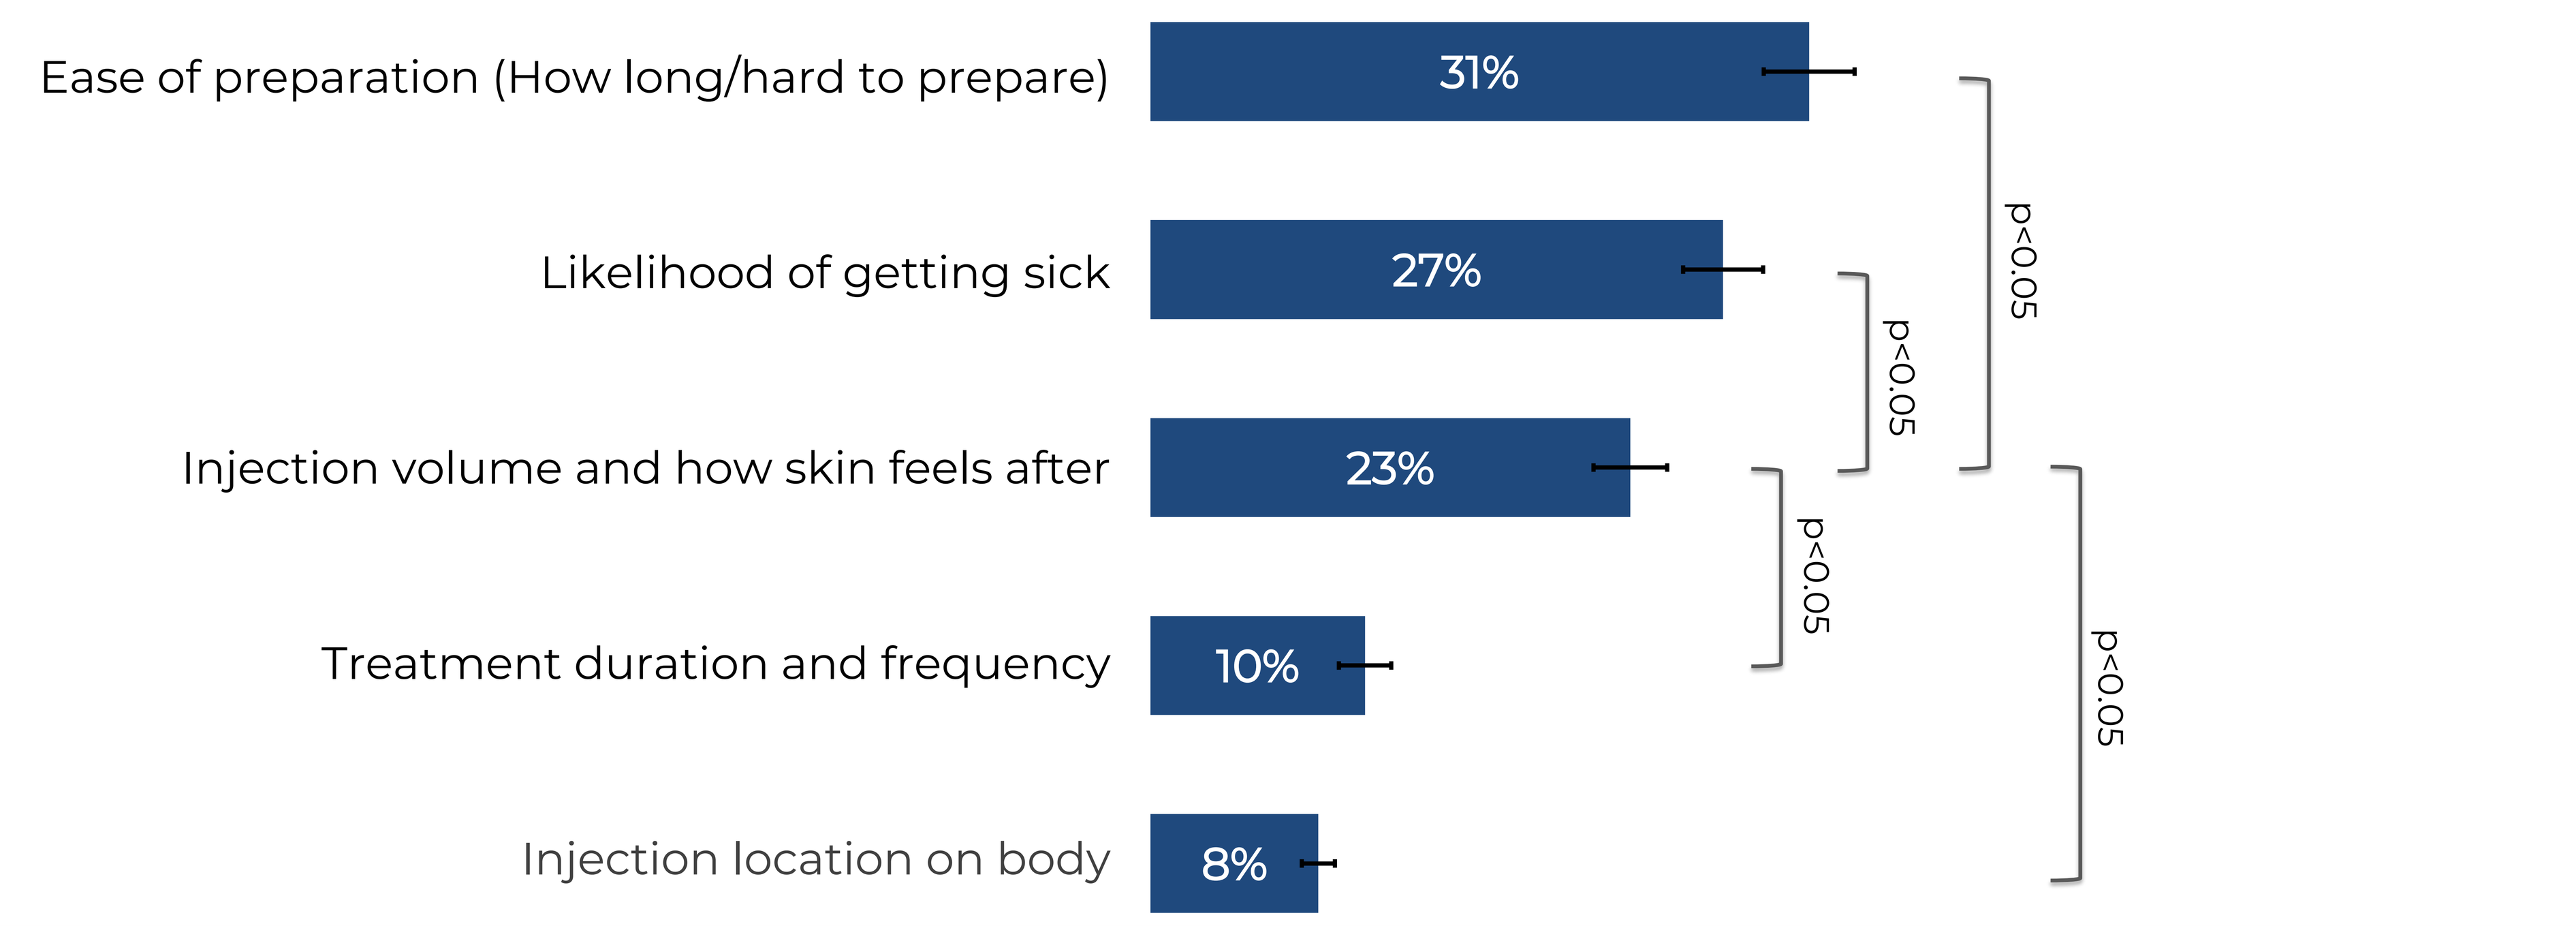
Ease of preparation (How long/hard to prepare)
31%
Likelihood of getting sick
27%
Injection volume and how skin feels after
23%
Treatment duration and frequency
10%
Injection location on body
8%
p<0.05
p<0.05
p<0.05
p<0.05

Figure 3. Self-reported confidence in ability to administer SCIg infusions

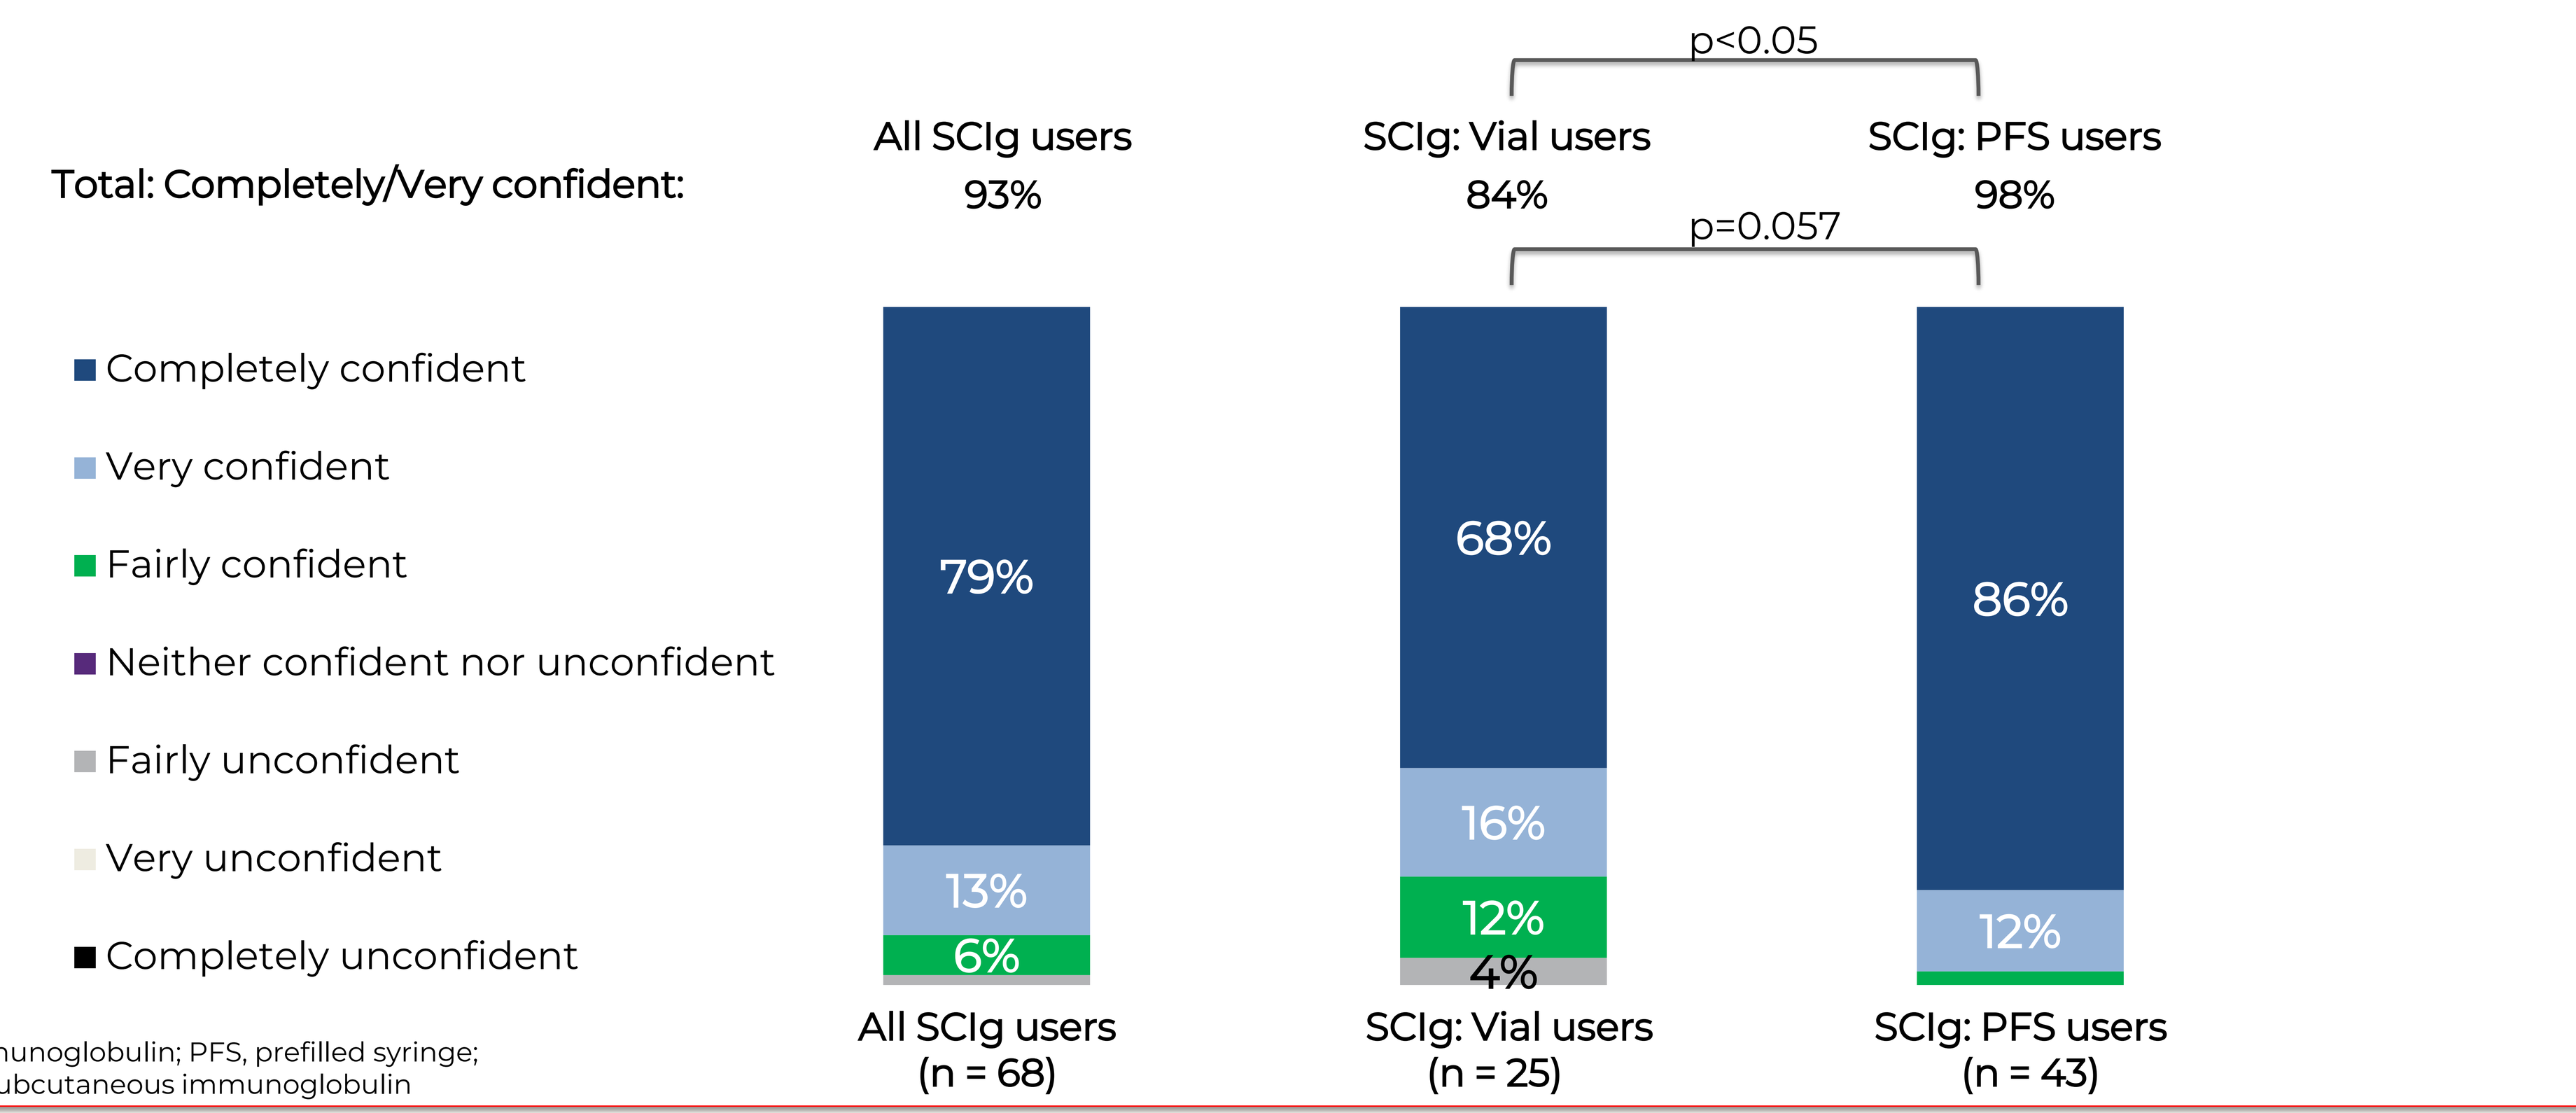
Total: Completely/Very confident:
All SCIg users 93%
SCIg: Vial users 84%
SCIg: PFS users 98%
p<0.05
p=0.057
Completely confident
Very confident
Fairly confident
Neither confident nor unconfident
Fairly unconfident
Very unconfident
Completely unconfident
79%
13%
6%
68%
16%
12%
4%
86%
12%
All SCIg users (n = 68)
SCIg: Vial users (n = 25)
SCIg: PFS users (n = 43)

Ig, immunoglobulin; PFS, prefilled syringe; SCIg, subcutaneous immunoglobulin

Figure 4. Self-reported 'feeling better' for vial and PFS users at various timepoints following their SCIg infusion

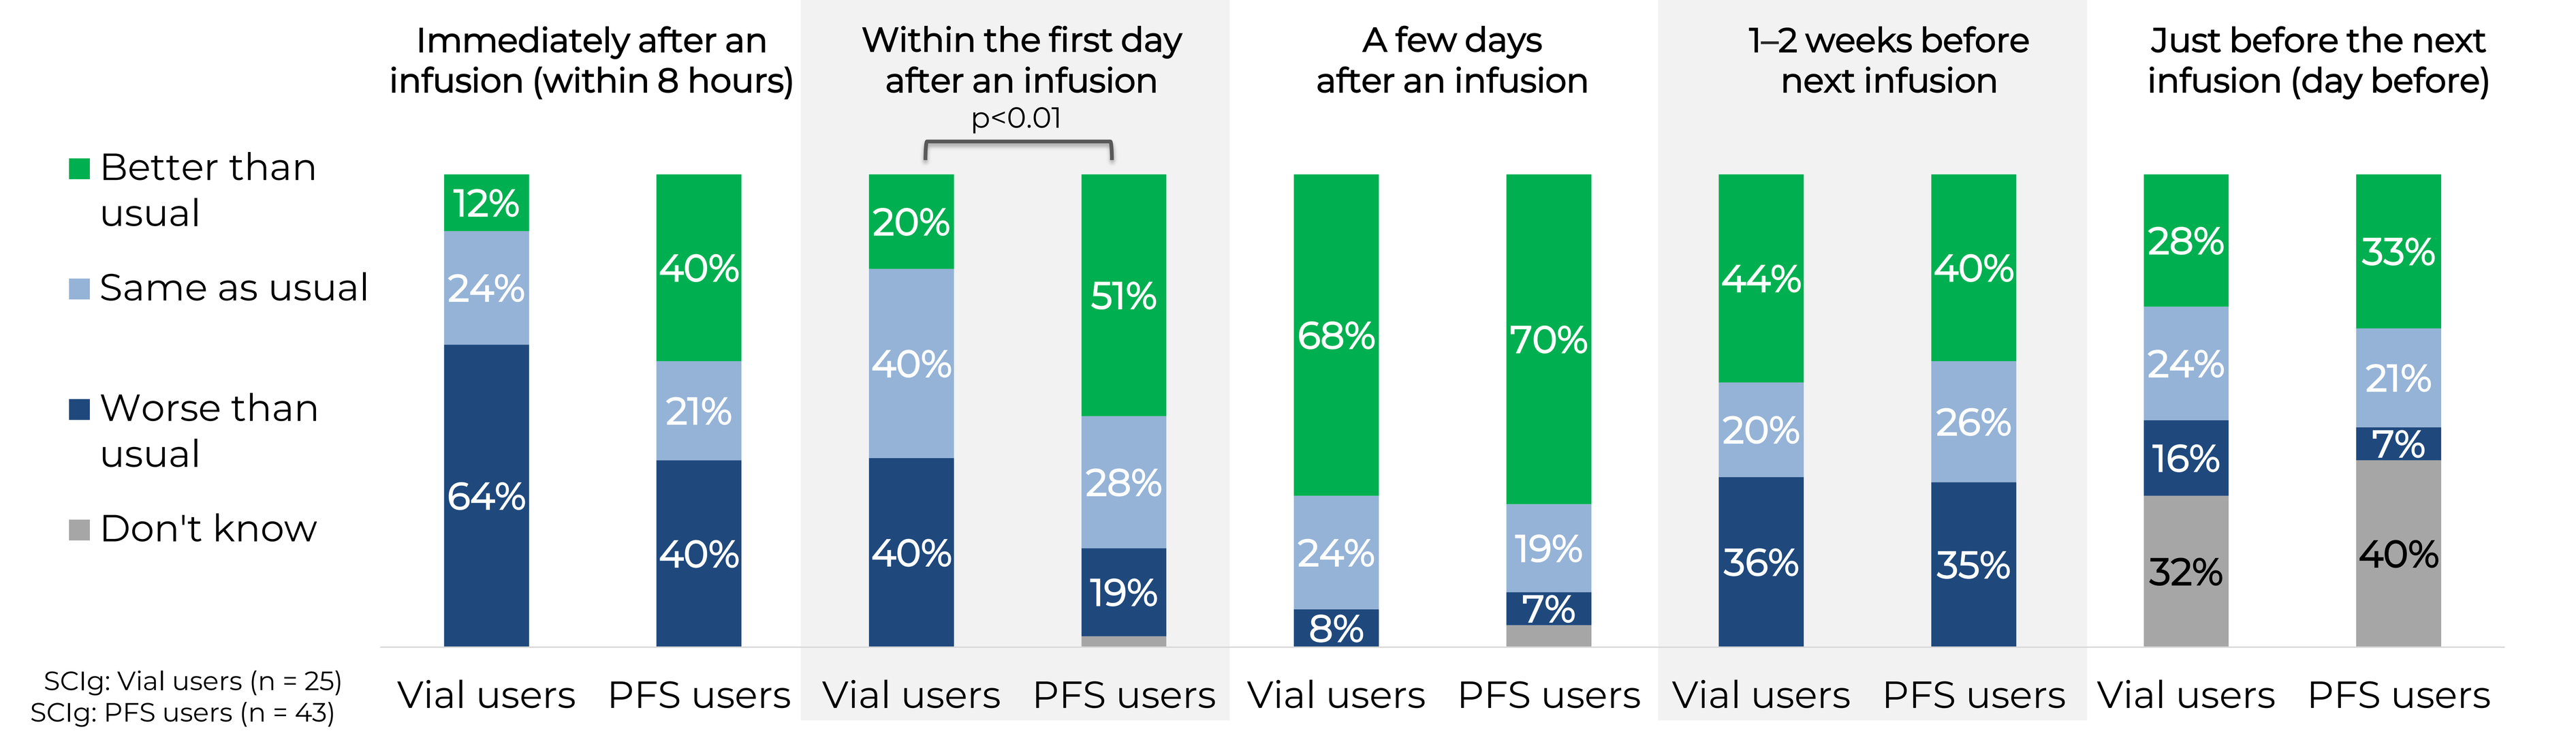
Immediately after an infusion (within 8 hours)
Within the first day after an infusion
A few days after an infusion
1–2 weeks before next infusion
Just before the next infusion (day before)
p<0.01
Better than usual
Same as usual
Worse than usual
Don't know
12%
24%
64%
40%
21%
40%
20%
40%
40%
51%
28%
19%
68%
24%
8%
70%
19%
7%
44%
20%
36%
40%
26%
35%
28%
24%
16%
32%
33%
21%
7%
40%
SCIg: Vial users (n = 25)
SCIg: PFS users (n = 43)
Vial users
PFS users
Vial users
PFS users
Vial users
PFS users
Vial users
PFS users
Vial users
PFS users

Ig, immunoglobulin; PFS, prefilled syringe; SCIg, subcutaneous immunoglobulin

References:

For further information please contact:
medinfoNA@cslbehring.com

QR

Acknowledgments: The original poster was developed with support from Bryter. Editorial support for the development of this encore poster was provided by Bioscript Group, UK.

Disclosures: **BK, JB, LU, SW,** and **PR** are employed at CSL and hold stocks at CSL. **SJC, NH,** and **BG** are full-time employees of Bryter Inc, and **BG** holds shares in its parent company Bryter Ltd.

This poster was previously presented at the 2026 Peripheral Nerve Society (PNS) Annual Meeting, Maastricht, The Netherlands, June 13–16, 2026.

Funding: This study was funded by CSL.

Presented at the 2026 American Association of Neuromuscular & Electrodiagnostic Medicine (AANEM) Annual Meeting, Orlando, Florida, USA, September 29–October 2, 2026.